Evaluation of non-gene edited allogeneic "off-the-shelf" Vδ1 γδ CAR T cells targeting CD20 for B cell malignancies

Aruna Azameera, Gauri Lamture, Taylor Barca, Alex Teague, Melinda Au, Jonathan Wong, Smitha Rao Yelaguli Gundurao, Amy Doan, Marissa Herrman, Swapna Panuganti, Arun Bhat, Blake T. Aftab, Kevin P. Nishimoto
Adicet Bio, Inc. Menlo Park, CA 94025

BACKGROUND

High clinical response rates have been observed with αβ CAR T therapies, but opportunities for improvement remain[1]. Off-the-shelf, allogeneic CAR T cell immunotherapies offer potential for immediately accessible cell therapies for patients. Strategies for investigating alternative cytotoxic effector cells with intrinsic tumoricidal activity, like γδ T cells, may improve depth and breadth of CAR T responses. Tumor targeting by allogeneic γδ CAR T therapy is complemented by innate and adaptive mechanisms[2]. ADI-001 is an allogeneic CD20-targeted γδ CAR T cell therapy currently being evaluated in patients with B cell lymphomas (NCT04735471)[3].

OBJECTIVE

To characterize the function and phenotype of ADI-001, a non-gene edited allogeneic Vδ1 γδ CAR T cell therapy targeting CD20, designed for the treatment of patients with B cell lymphomas.

Manufacturing of allogeneic CD20 CAR Vδ1 γδ T cells

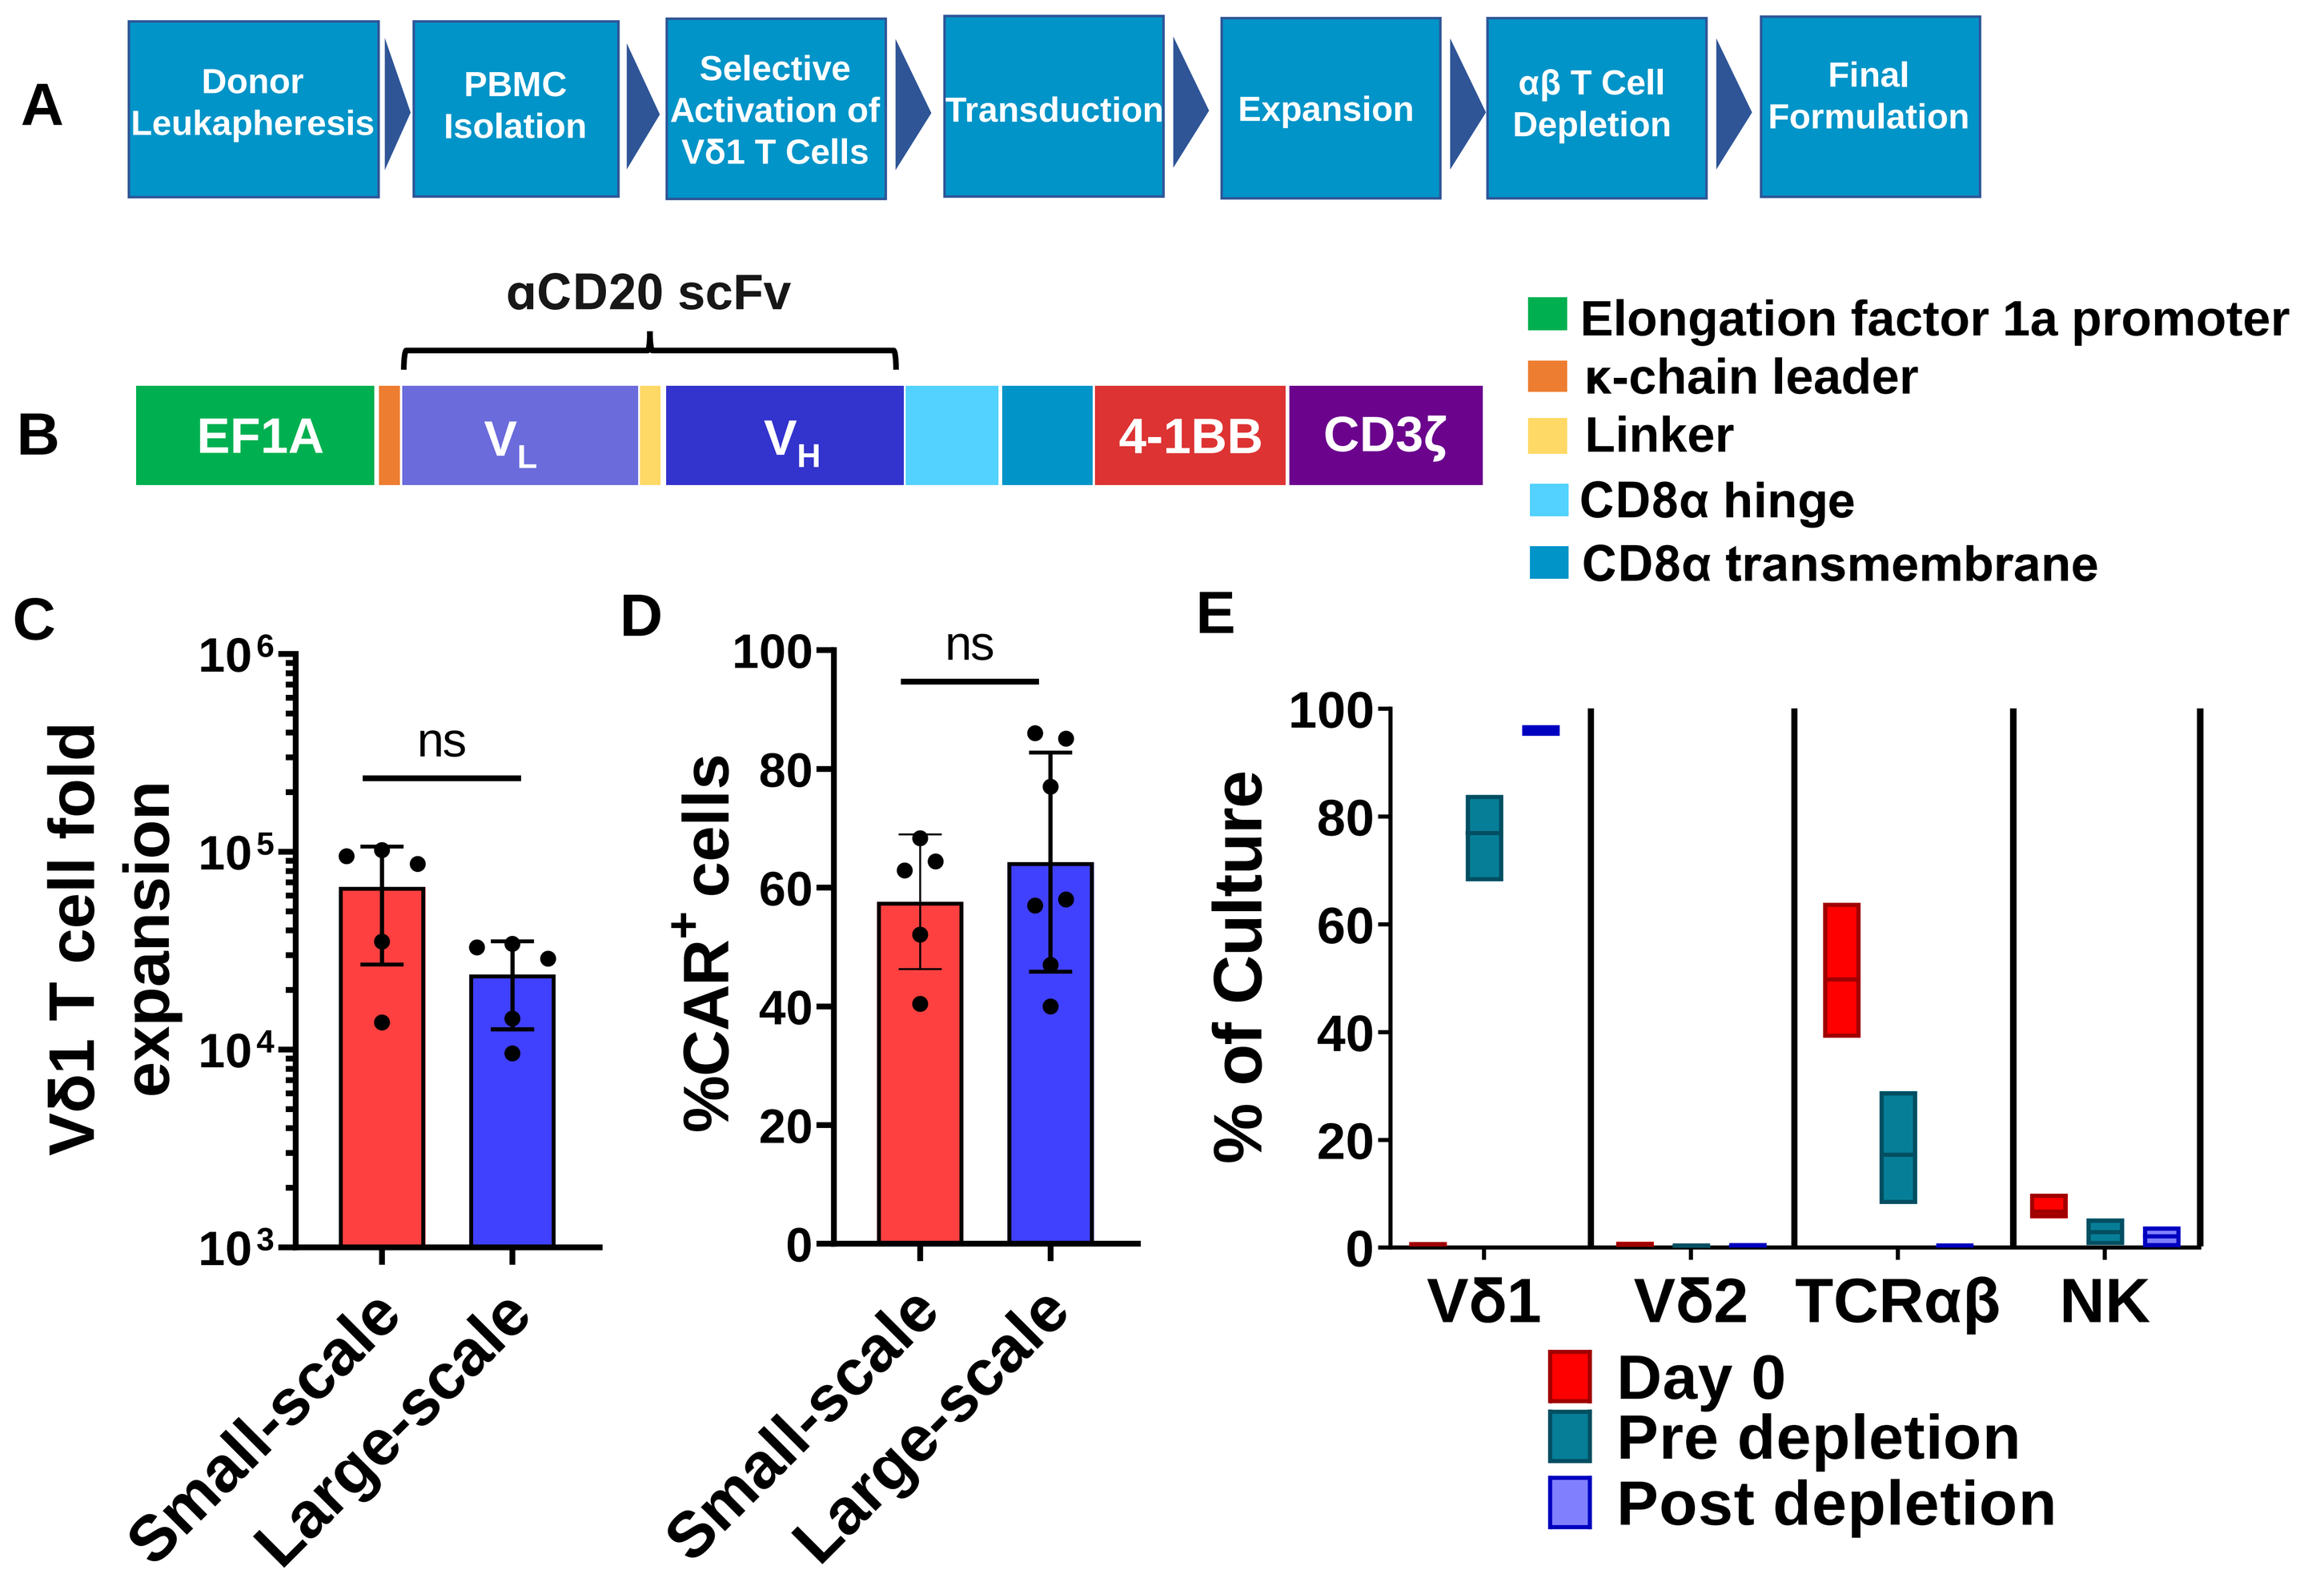
A
Donor Leukapheresis
PBMC Isolation
Selective Activation of Vδ1 T Cells
Transduction
Expansion
αβ T Cell Depletion
Final Formulation
B
αCD20 scFv
EF1A
VL
VH
4-1BB
CD3ζ
Elongation factor 1a promoter
κ-chain leader
Linker
CD8α hinge
CD8α transmembrane
C
Vδ1 T cell fold expansion
ns
Small-scale
Large-scale
D
%CAR+ cells
ns
Small-scale
Large-scale
E
% of Culture
Vδ1
Vδ2
TCRαβ
NK
Day 0
Pre depletion
Post depletion

Figure 1. Selective activation and expansion of Vδ1 γδ T cells using agonistic mAb from healthy donor-derived PBMCs. **(A)** Flow chart highlighting the key steps in the manufacturing of allogeneic CD20 CAR$^+$ Vδ1 γδ T cells. **(B)** Schematic diagram of the second generation CD20 CAR. **(C)** CD20 CAR$^+$ Vδ1 γδ T cell manufacturing process resulted in a substantial fold-expansion of Vδ1 γδ T cells. **(D)** Average percentage of Vδ1 γδ T cells expressing the CD20 CAR from manufacturing runs as measured by flow cytometry. **(E)** % Cell composition throughout expansion of CD20 CAR$^+$ Vδ1 T cell products derived from 4 different donors analyzed by flow cytometry. Paired t-test was used to assess statistical significance.

Predominant naïve-like T cell memory phenotype, NKR and chemokine receptor expression in CD20 CAR Vδ1 γδ T cells

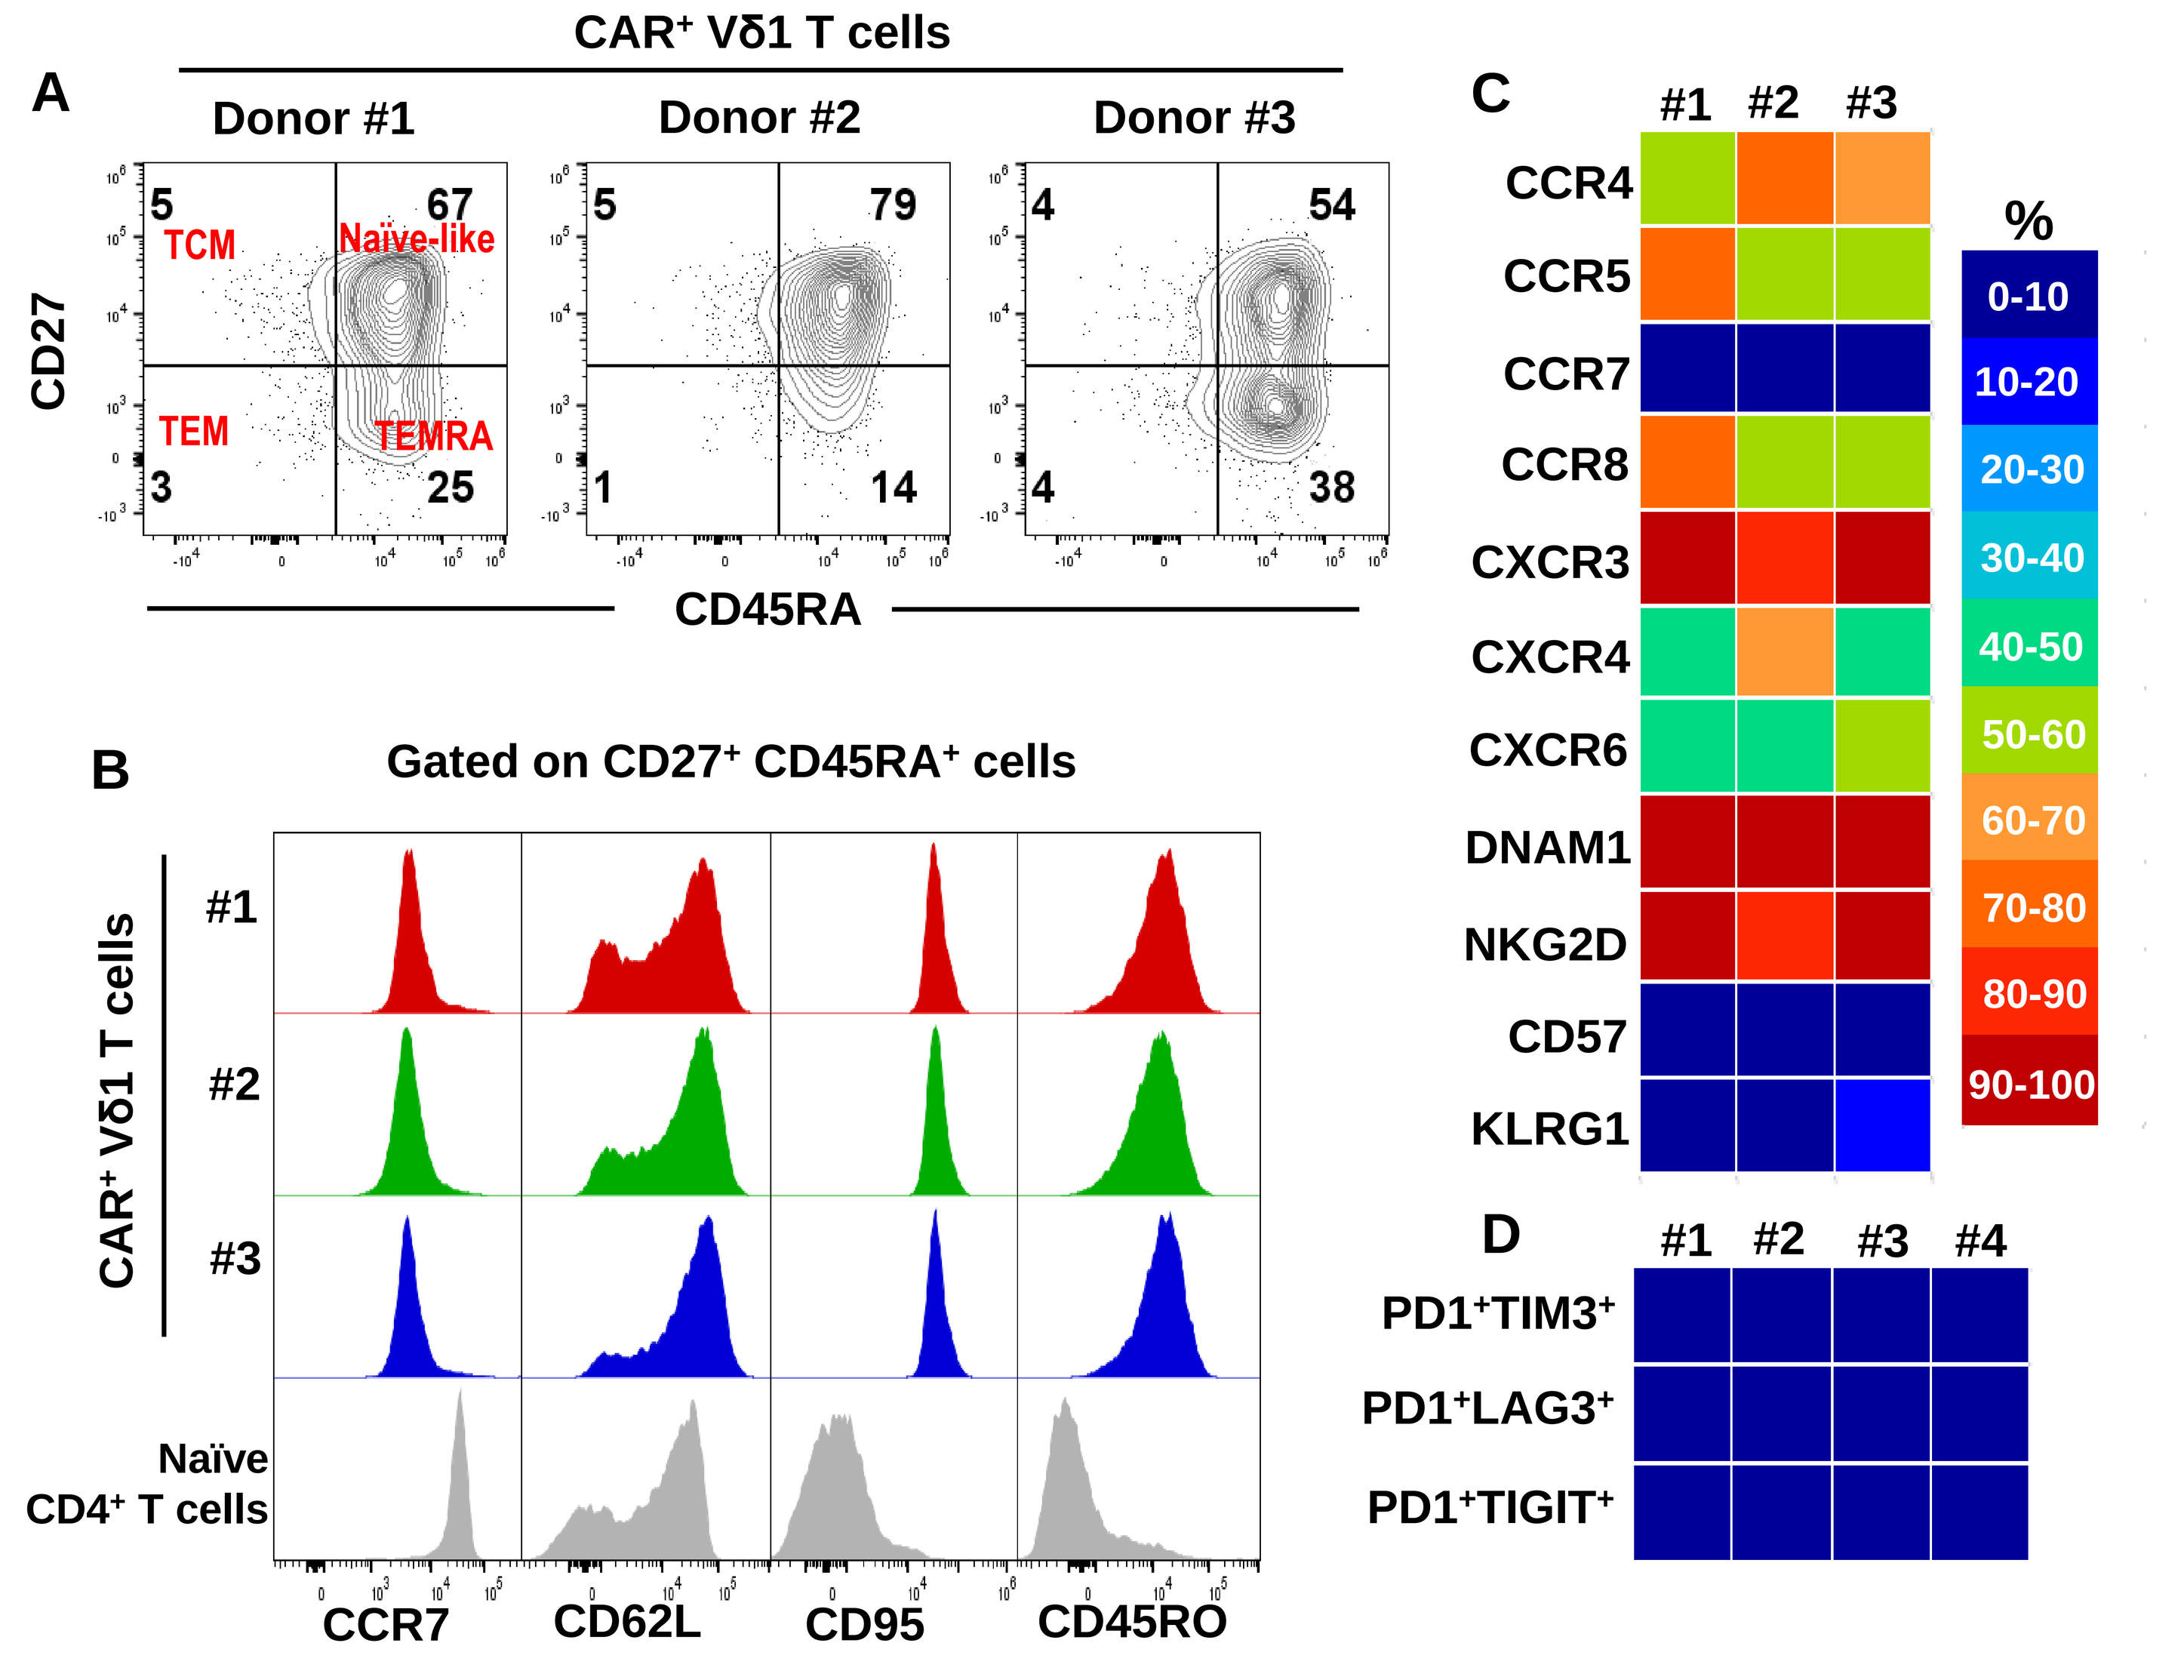
A
CAR+ Vδ1 T cells
Donor #1
Donor #2
Donor #3
TCM
Naïve-like
TEM
TEMRA
CD27
CD45RA
B
Gated on CD27+ CD45RA+ cells
CAR+ Vδ1 T cells
#1
#2
#3
Naïve CD4+ T cells
CCR7
CD62L
CD95
CD45RO
C
#1 #2 #3
CCR4
CCR5
CCR7
CCR8
CXCR3
CXCR4
CXCR6
DNAM1
NKG2D
CD57
KLRG1
%
0-10
10-20
20-30
30-40
40-50
50-60
60-70
70-80
80-90
90-100
D
#1 #2 #3 #4
PD1+TIM3+
PD1+LAG3+
PD1+TIGIT+

Figure 2. (A and B) Majority of CAR$^+$ Vδ1 γδ T cells exhibited a naïve-like T cell memory phenotype assessed by flow cytometry. **(C)** Heatmap showing percentages of CAR$^+$ Vδ1 γδ T cells in CD20 CAR$^+$ Vδ1 γδ T cell products that express multiple chemokine receptors, natural killer (NK) cell receptors, and terminal differentiation markers. **(D)** Heatmap showing percentages of CAR$^+$ Vδ1 γδ T cells in CD20 CAR$^+$ Vδ1 γδ T cell products that co-express PD-1 and another co-inhibitory receptor.

CD20 CAR Vδ1 γδ T cells exhibit potent cytotoxicity and proliferation against B cell lymphoma cell lines

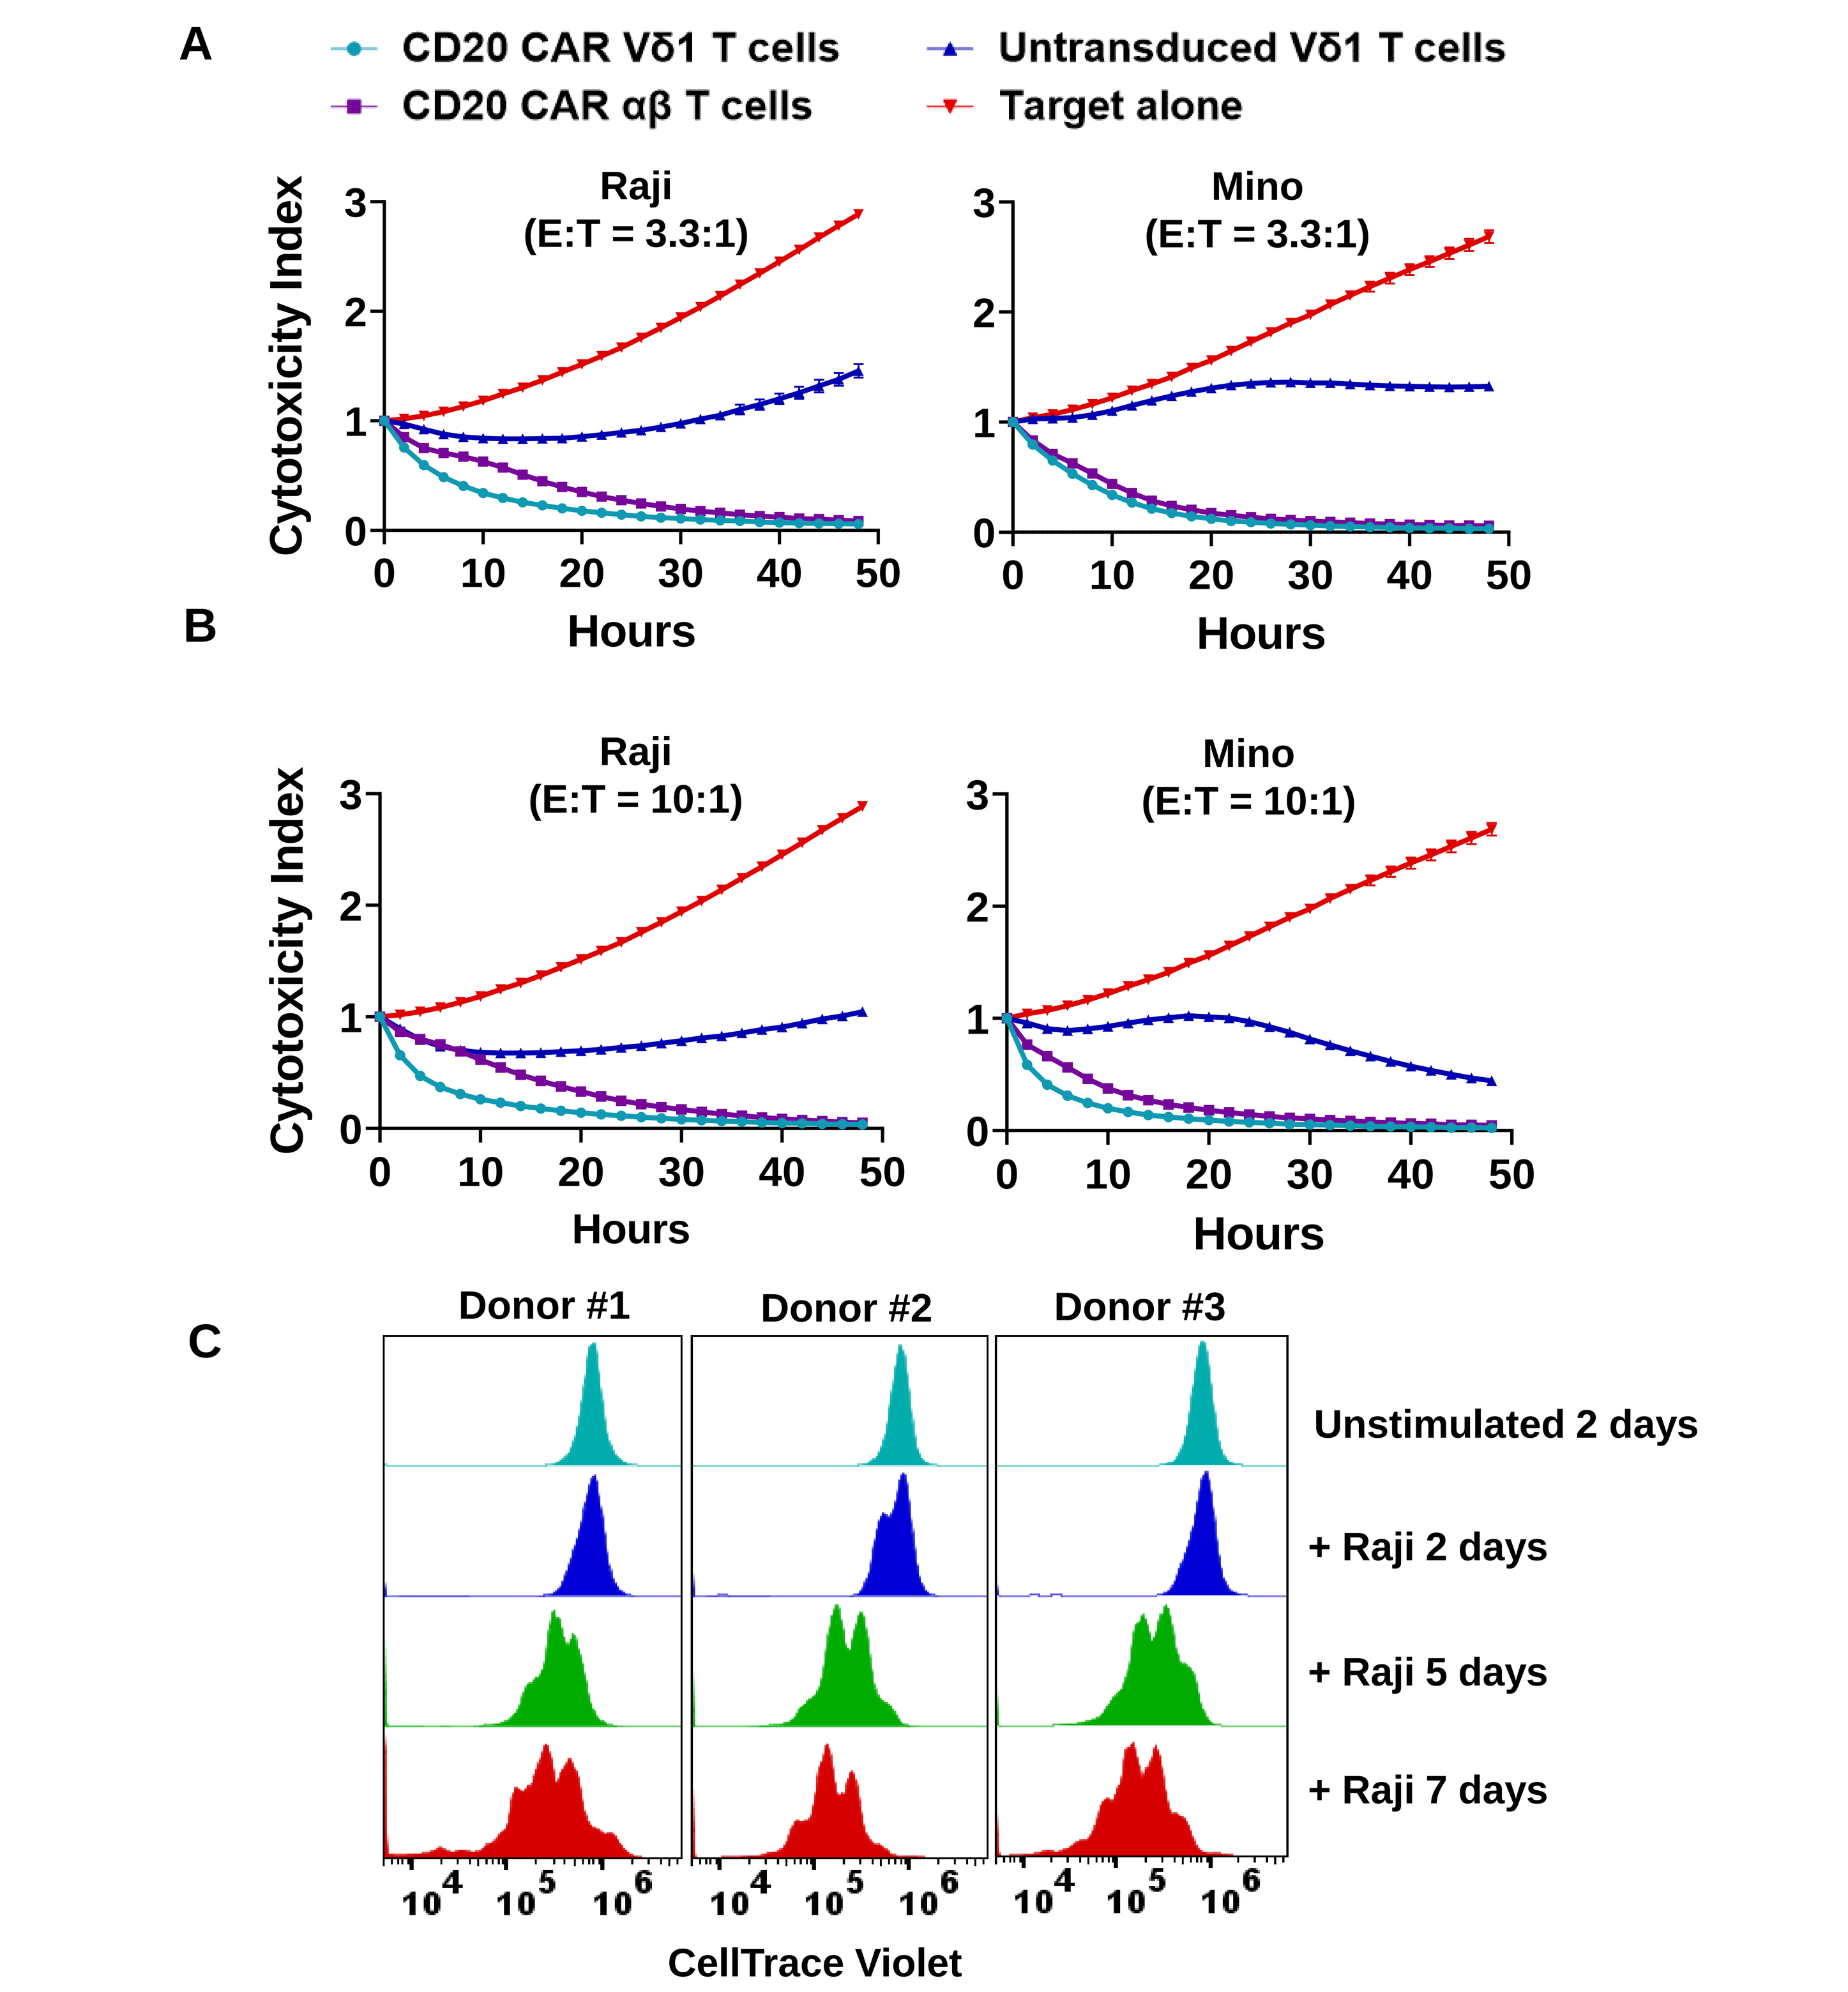
A
CD20 CAR Vδ1 T cells
CD20 CAR αβ T cells
Untransduced Vδ1 T cells
Target alone
Raji (E:T = 3.3:1)
Mino (E:T = 3.3:1)
Cytotoxicity Index
Hours
B
Raji (E:T = 10:1)
Mino (E:T = 10:1)
C
Donor #1
Donor #2
Donor #3
Unstimulated 2 days
+ Raji 2 days
+ Raji 5 days
+ Raji 7 days
CellTrace Violet

Figure 3. (A and B) Cytotoxic potentials of untransduced Vδ1 γδ T cells, CD20 CAR-transduced Vδ1 γδ T cells (71.5% CAR$^+$), and CD20 CAR-transduced αβ T cells (68% CAR$^+$), all derived from the same healthy donor, were evaluated against CD20$^+$ target cells in a 48-hour Incucyte Immune Cell Killing Assay, in which T cells were co-cultured with NucR-expressing Raji or Mino target cells at E:T ratios of 3.3:1 and 10:1. **(C)** Proliferative potential of CD20 CAR$^+$ Vδ1 γδ T cells following three rounds of target antigen exposure in a 7-day culture period.

CD20 CAR Vδ1 γδ T cells significantly inhibit *in vivo* tumor growth in B cell lymphoma xenografts

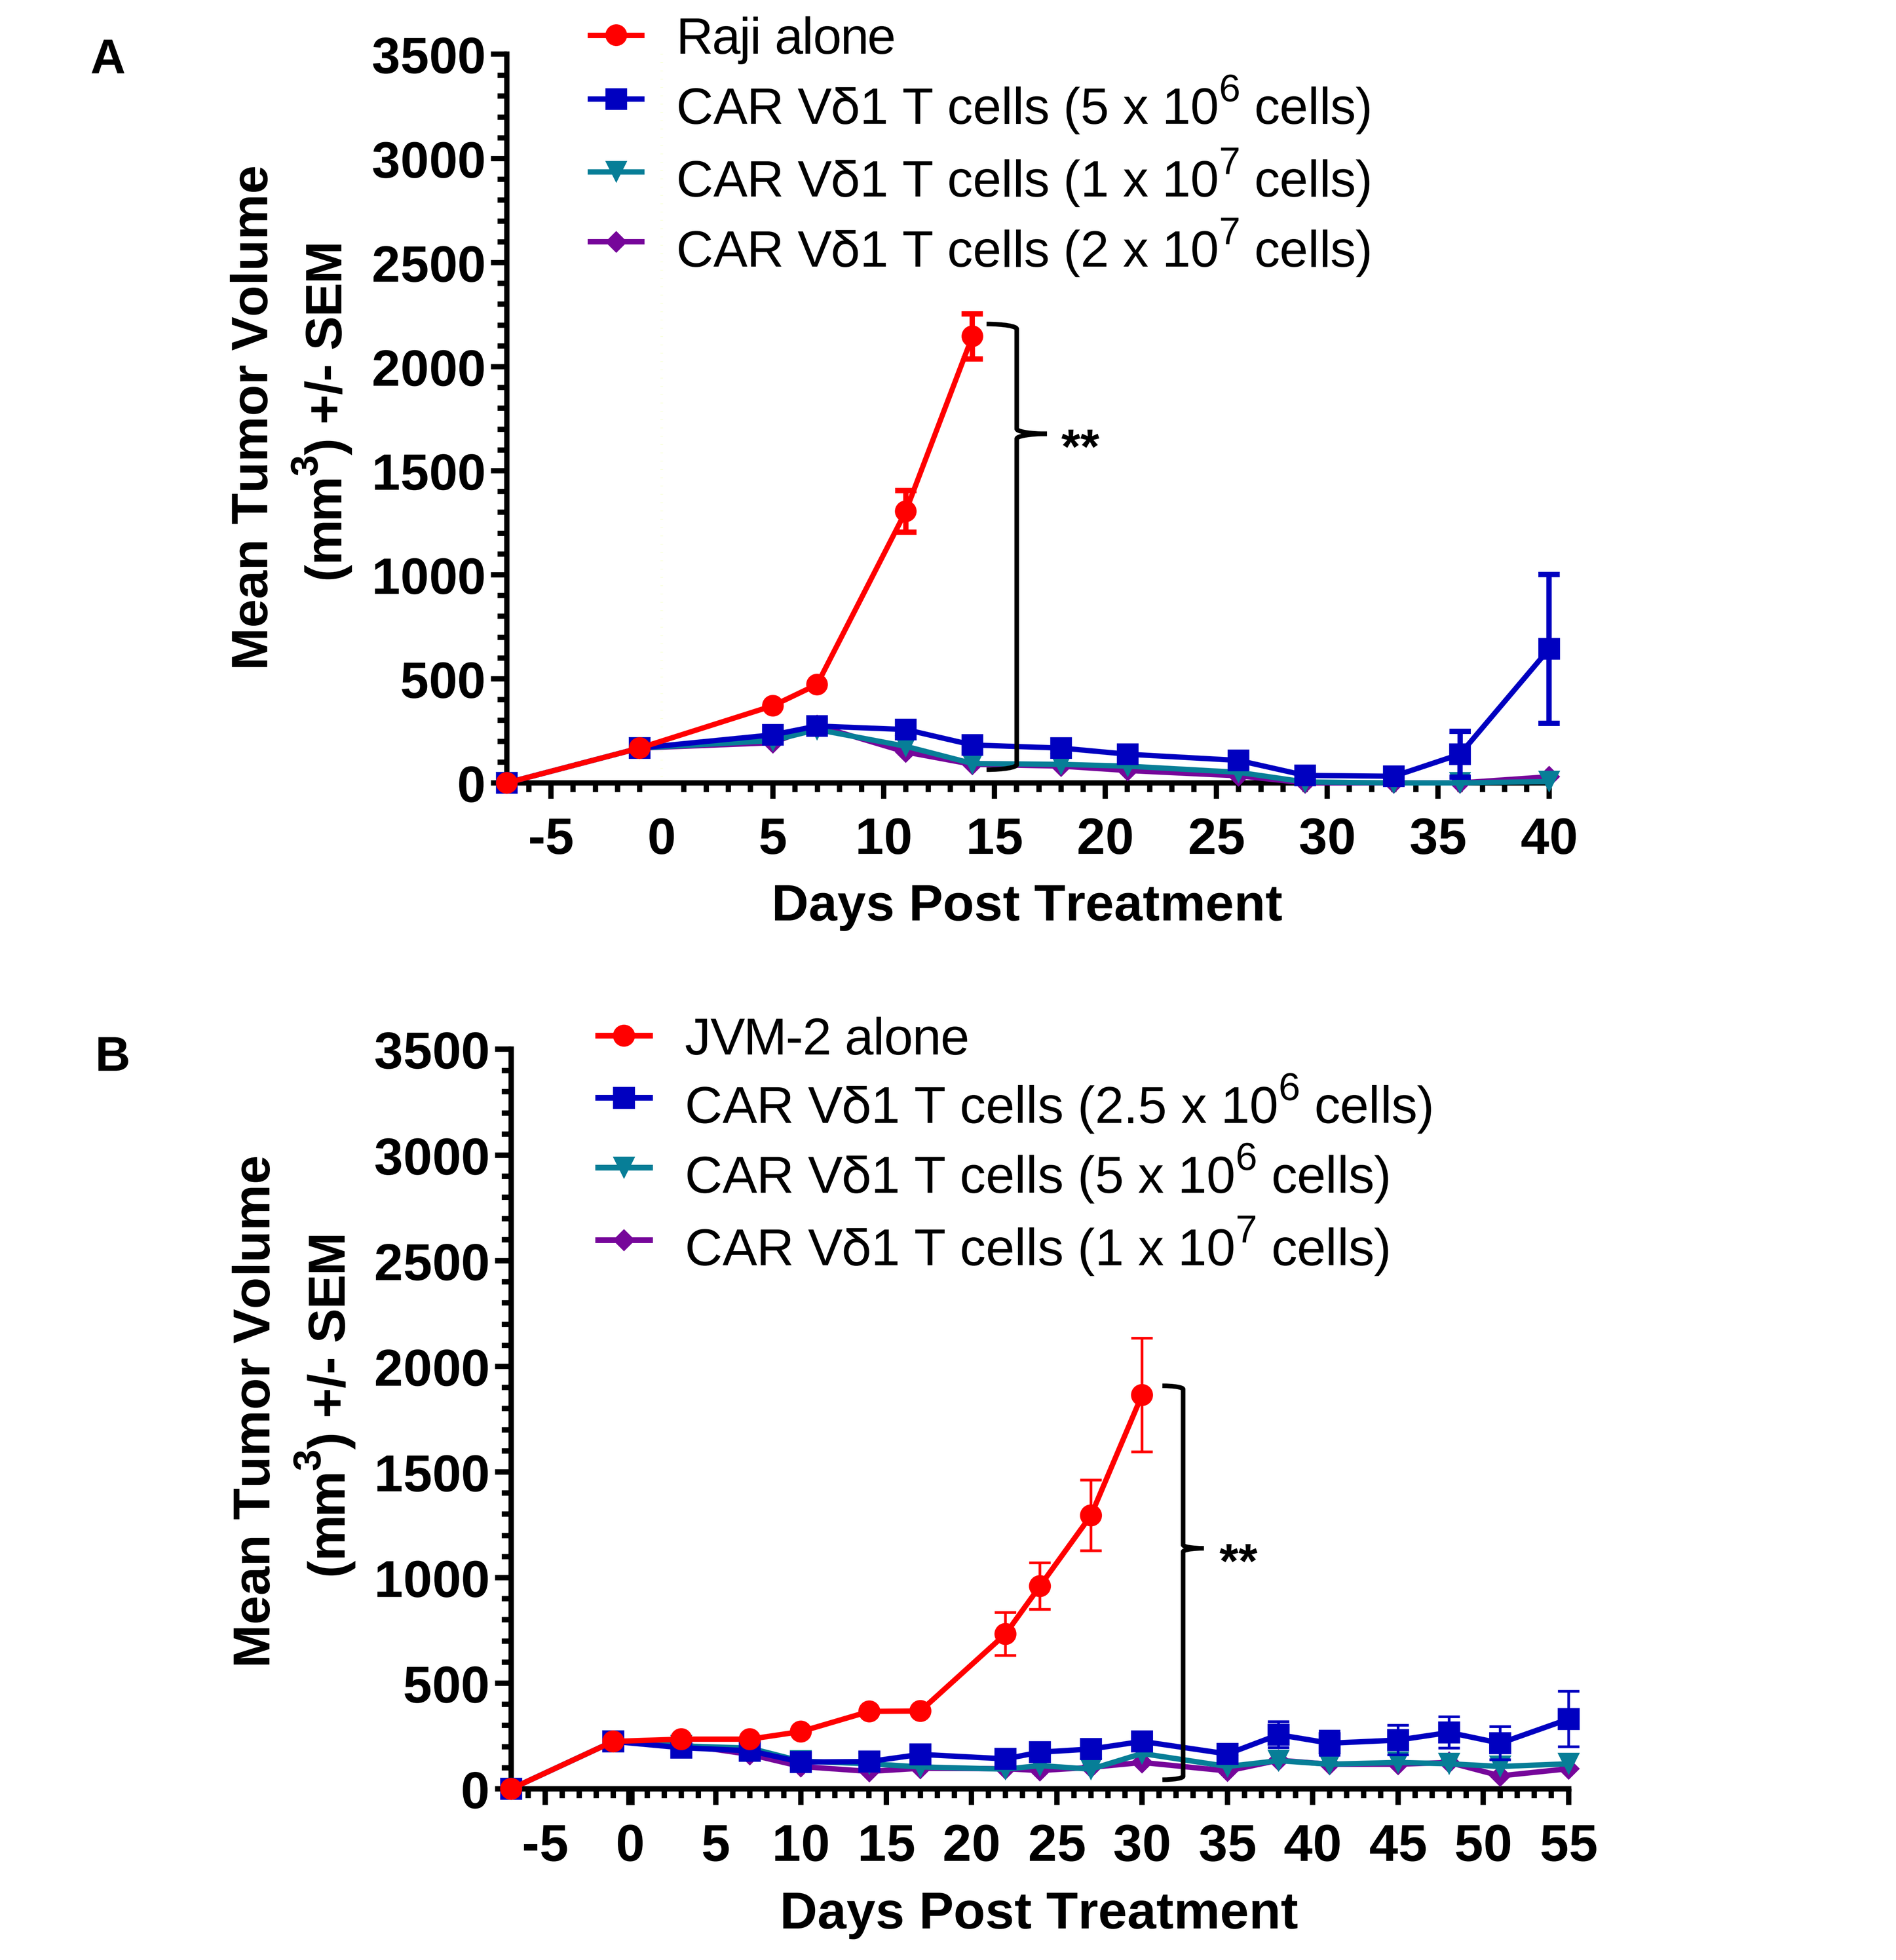
A
Raji alone
CAR Vδ1 T cells (5 x 10^6 cells)
CAR Vδ1 T cells (1 x 10^7 cells)
CAR Vδ1 T cells (2 x 10^7 cells)
Mean Tumor Volume (mm^3) +/- SEM
Days Post Treatment
**
B
JVM-2 alone
CAR Vδ1 T cells (2.5 x 10^6 cells)
CAR Vδ1 T cells (5 x 10^6 cells)
CAR Vδ1 T cells (1 x 10^7 cells)
Mean Tumor Volume (mm^3) +/- SEM
Days Post Treatment
**

Figure 4. *In vivo* efficacy of three different doses of viable CD20 CAR$^+$ Vδ1 γδ T cells in combination with 13,000 IU IL-2 in **(A)** SC Raji Burkitt Lymphoma and **(B)** SC JVM-2 Mantle Cell Lymphoma model in NSG mice (n = 5 per group). Kruskal-Wallis test was used to assess statistical significance among the groups (***P*-value < 0.01).

Innate and adaptive immune contributions for Vδ1 γδ CAR T cell targeting of B cell lymphomas

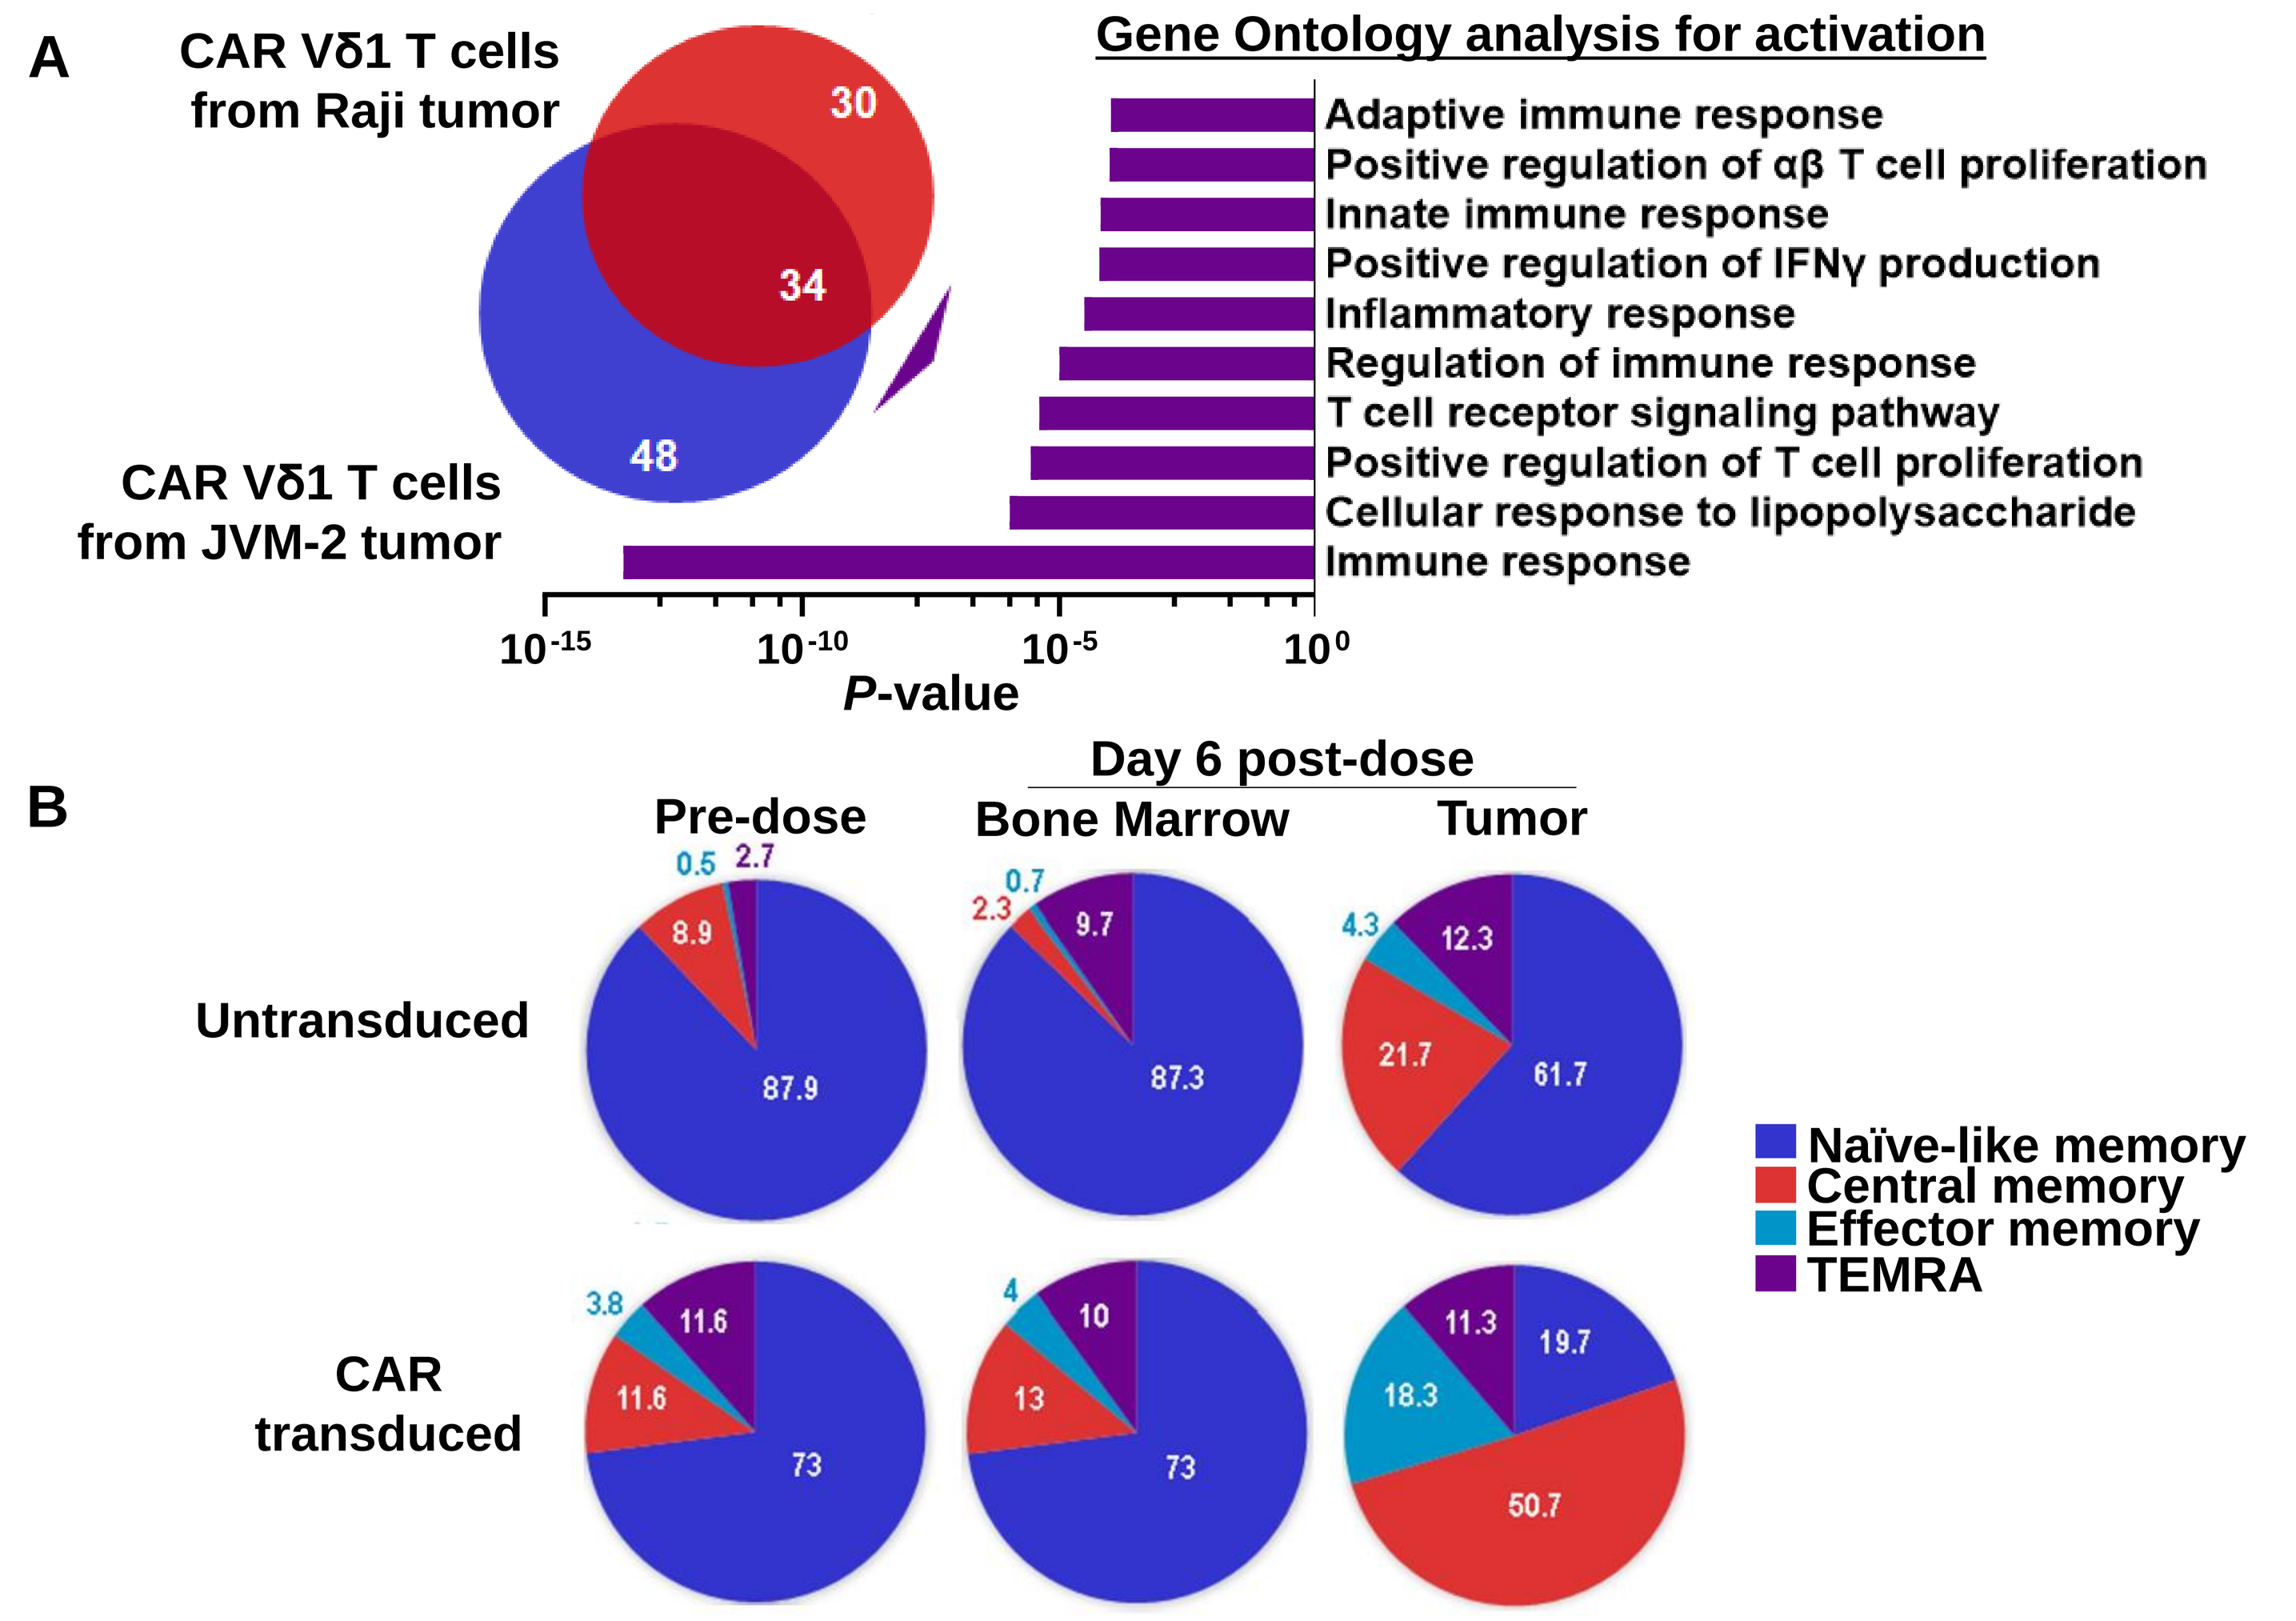
A
CAR Vδ1 T cells from Raji tumor
CAR Vδ1 T cells from JVM-2 tumor
30
34
48
Gene Ontology analysis for activation
Adaptive immune response
Positive regulation of αβ T cell proliferation
Innate immune response
Positive regulation of IFNγ production
Inflammatory response
Regulation of immune response
T cell receptor signaling pathway
Positive regulation of T cell proliferation
Cellular response to lipopolysaccharide
Immune response
P-value
B
Day 6 post-dose
Pre-dose
Bone Marrow
Tumor
Untransduced
CAR transduced
Naïve-like memory
Central memory
Effector memory
TEMRA

Figure 5. (A) Venn diagrams of differentially expressed genes (DEGs) from Raji (red circle) and JVM-2 (blue circle) tumors treated with CD20 CAR$^+$ Vδ1 γδ T cells (day 6 post-dose). The overlapping area represents DEGs that are shared between the two tumor models. Gene expression were quantitated using the Nanostring nCounter® CAR T Characterization and Human Immunology Panels. Gene Ontology analysis was performed using Database for Annotation, Visualization and Integrated Discovery (https://david.ncifcrf.gov/) to identify the biological pathways associated with DEGs that are unique to each tumor model and that are shared by the two tumor models **(B)** T cell memory phenotype analysis of untransduced and CD20 CAR$^+$ Vδ1 γδ T cells in bone marrow and tumor harvested from Raji tumor-bearing mice on day 6 post-treatment.

Non-edited Vδ1 γδ CAR T cells show decreased HvG potential compared to gene-edited platform variants

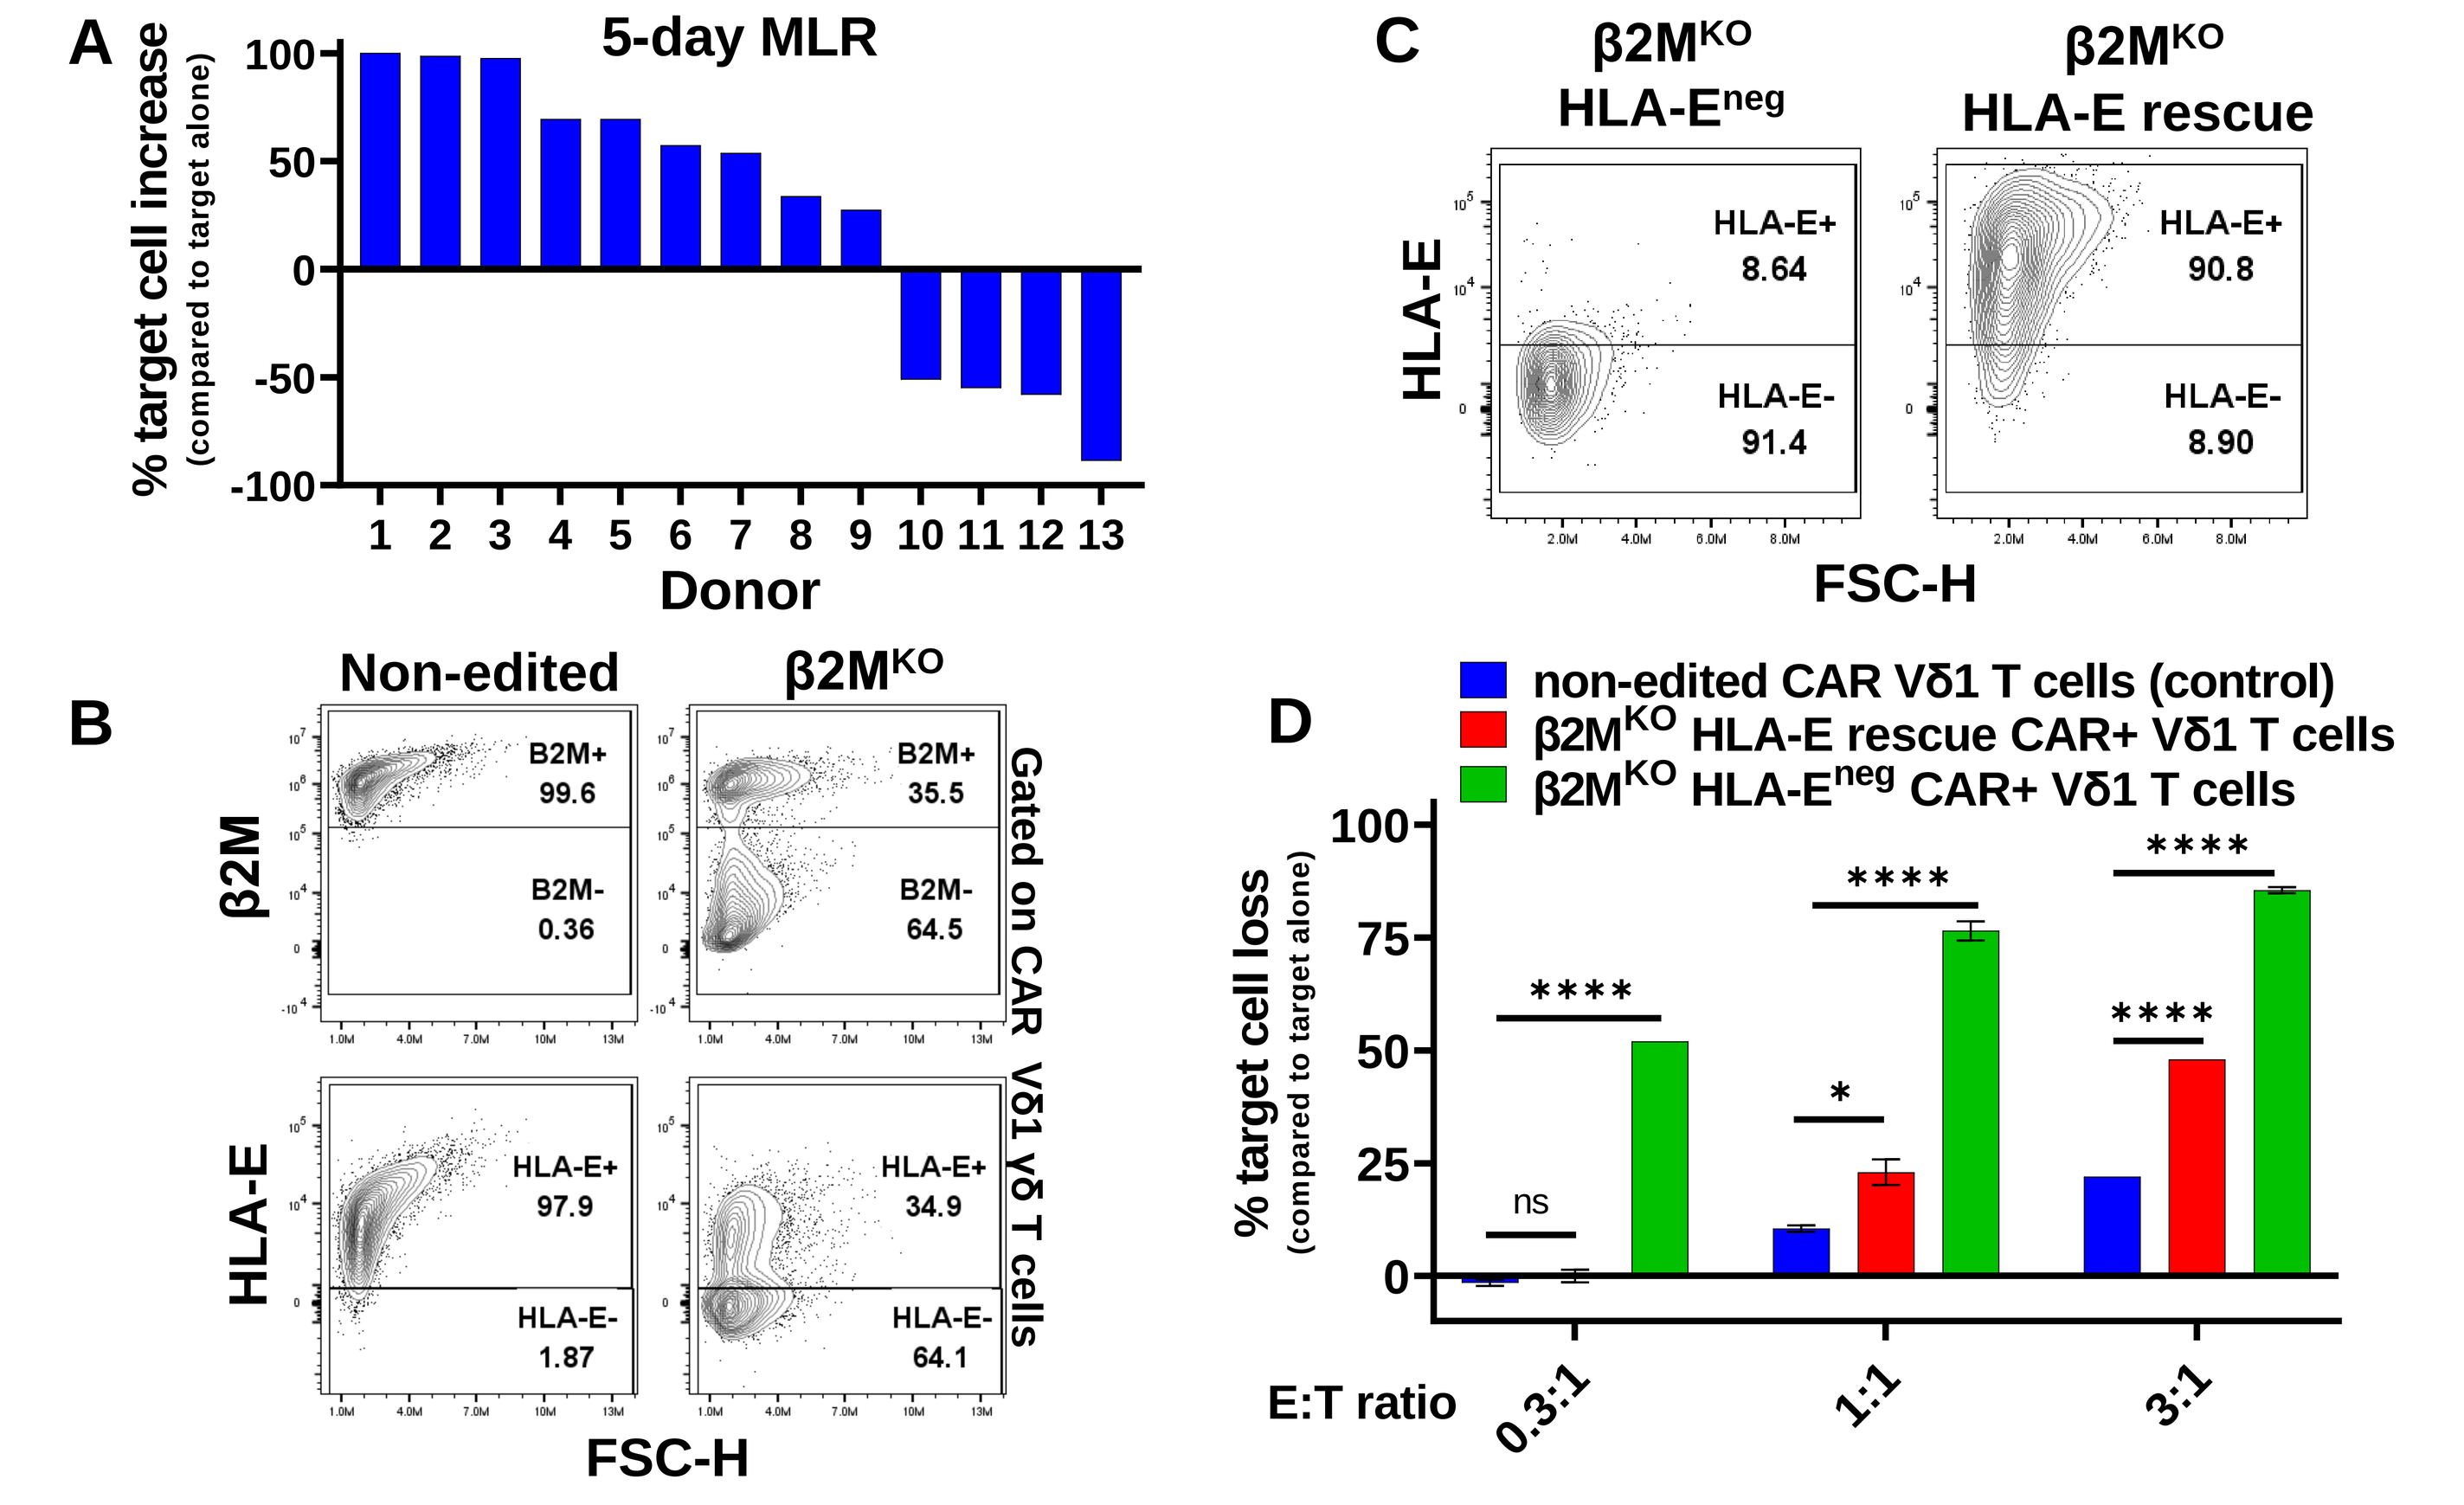
A
5-day MLR
% target cell increase (compared to target alone)
Donor
B
Non-edited
β2M^KO
β2M
HLA-E
FSC-H
Gated on CAR Vδ1 γδ T cells
C
β2M^KO HLA-E^neg
β2M^KO HLA-E rescue
HLA-E
FSC-H
D
non-edited CAR Vδ1 T cells (control)
β2M^KO HLA-E rescue CAR+ Vδ1 T cells
β2M^KO HLA-E^neg CAR+ Vδ1 T cells
% target cell loss (compared to target alone)
E:T ratio
0.3:1
1:1
3:1

Figure 6. (A) Assessment of allo-susceptibility using enriched primary lymphocytes from allogeneic PBMC donors (responders) against non-edited CD20 CAR Vδ1 γδ T cells (targets) in a 5-day MLR. **(B)** Generation of β2M KO CAR$^+$ Vδ1 γδ T cells was assessed using flow cytometry. **(C)** β2M KO with single-chain HLA-E rescue expression was assessed using flow cytometry after cell sorting. **(D)** NK cell allo-susceptibility against non-gene edited CD20 CAR$^+$ Vδ1 γδ T cells was compared to β2M KO with and without single chain HLA-E rescued CD20 CAR$^+$ Vδ1 γδ T cells. Enriched, activated, and expanded allogeneic NK cells (responders) were co-cultured with CFSE-labeled CD20 CAR$^+$ Vδ1 γδ T cells (targets) for 18-hrs and cell killing was assessed using flow cytometry.
(*****P*-value < 0.001, **P*-value < 0.05, NS = not significant).

CONCLUSIONS

- PBMC-derived Vδ1 γδ T cells were successfully activated, expanded, and genetically engineered using established manufacturing processes.
- ADI-001 demonstrated a predominantly naïve-like T cell memory phenotype and expressed multiple chemokine and natural killer cell receptors.
- ADI-001 exhibited robust *in vitro* and *in vivo* tumor growth inhibition in multiple human lymphoma cell lines.
- Adaptive and innate mechanisms contribute to the anti-tumor activity of ADI-001
- Vδ1 γδ CAR T cells may be relatively resilient to host vs graft rejection when compared to gene-edited approaches ($\beta 2M^{KO}$ with or without HLA-E overexpression), based on mixed lymphocyte reactions with mismatched allogeneic PBMC and NK donor lymphocytes.
- These findings demonstrate preclinical proof-of-concept for ADI-001 as an allogeneic CAR-T therapy. A phase 1 trial using ADI-001 to treat R/R B cell NHL patients is currently under investigation (NCT04735471)[3].

References

1. McCreedy BJ, Senyukov VV, Nguyen KT. Off the shelf T cell therapies for hematologic malignancies. *Best Pract Res Clin Haematol* 2018; **31**: 166-175.
2. Bonneville M, O'Brien RL, Born WK. γδ T cell effector functions: a blend of innate programming and acquired plasticity. *Nat Rev Immunol* 2010; **10**: 467-478.
3. A Study of ADI-001 in B Cell Malignancies (GLEAN-1). *https://www.clinicaltrials.gov/ct2/show/NCT04735471*