A Novel γδ T Cell Product Targeting CD20 for the Treatment of B Cell Malignancies

Jason Romero, Marissa Herrman, Kevin Nishimoto, Bernadette Dahlin, Chris Chavez, Taylor Barca, Charles Feathers, Max Lee, Praveen Tayakuniyil, Arnaud Colantonio, Zili An, Frank Jing, Stewart Abbot and Daulet Satpayev

Adicet Bio Inc., Menlo Park, CA 94025 US

Introduction

Wide-spread adoption of autologous cell therapies continues to be challenged by manufacturing difficulties, availability, safety, consistency and cost of production. In order to overcome many of these challenges, we are developing an alternative approach employing allogeneic gamma delta (γδ) T cells to create a universal T cell therapy for the treatment of cancer. γδ T cells are a minor lymphocyte population in the circulation but can be found in larger numbers in tissues where they exhibit potent immune monitoring, anti-viral, anti-tumor and anti-microbial functions. γδ T cells can recognize pathogen stressed and transformed target cells in an HLA independent fashion and represent a functional "bridge" between innate and adaptive immunity thereby facilitating activation in an allogeneic setting without the concern of Graft versus Host Disease (GvHD) that challenges development of allogeneic αβ T cells.

Here we present a Vδ1 CAR T cell product targeting CD20 antigen for the treatment of B cell malignancies. Vδ1 T cells are the predominant tissue-associated γδ T cell subset in humans and are thought to recognize signs of cellular stress, including viral infection and transformation. In our process, Vδ1 cells were selectively expanded from peripheral blood of normal healthy donors and engineered with a 2nd generation CAR construct (4-1BBz) to create an allogeneic CAR T cell product. The level of expansion, purity and retention of excellent anti-tumor potency following cryopreservation support creation of a substantial number of doses (~1000 per production run) of well characterized, uniform product that is available in an "off-the-shelf" manner. CAR engineering substantially adds to the innate anti-lymphoma activity of these cells and potentiates the multi-receptor targeting of lymphoma cell lines.

We have demonstrated that in response to target cells, Vδ1 CD20 CAR T cells secrete effector cytokines, induce apoptosis and clear lymphoma cells in vitro, and these functions can be further potentiated by providing exogenous cytokines to support proliferation. When tested in vivo, Vδ1 CD20 CAR T cells demonstrated potent antitumor activity in both disseminated and subcutaneous models of B cell lymphoma without xenogeneic GvHD. We show that the 4-1BB co-stimulatory domain significantly improves the persistence of Vδ1 CAR T cells, and additional engineering may further optimize the cell product. Overall, these data show selectively expanded Vδ1 T cells represent a unique, safe and effective platform for therapeutic intervention in various cancers which warrants further clinical investigation of this CD20-targeted Vδ1 CAR T cell product drug candidate.

Manufacturing of Allogeneic Vδ1 CAR T Cells

Selection of Optimal Healthy Donors → Manufacturing Process < 4 weeks → Batch Sizes of 2E11 Cells

- Robust activation using a proprietary γδ TCR monoclonal antibody
- Viral transduction using γ-retroviral vector to introduce CAR transgene
- Large scale perfusion bioreactor-based expansion
- Excellent post cryopreservation viability and function
- ~1000 doses per batch; available "off-the-shelf"
- cGMP compliant manufacturing

CAR Construct: αCD20 scFv – CD8α H-TM – 4-1BB – CD3z

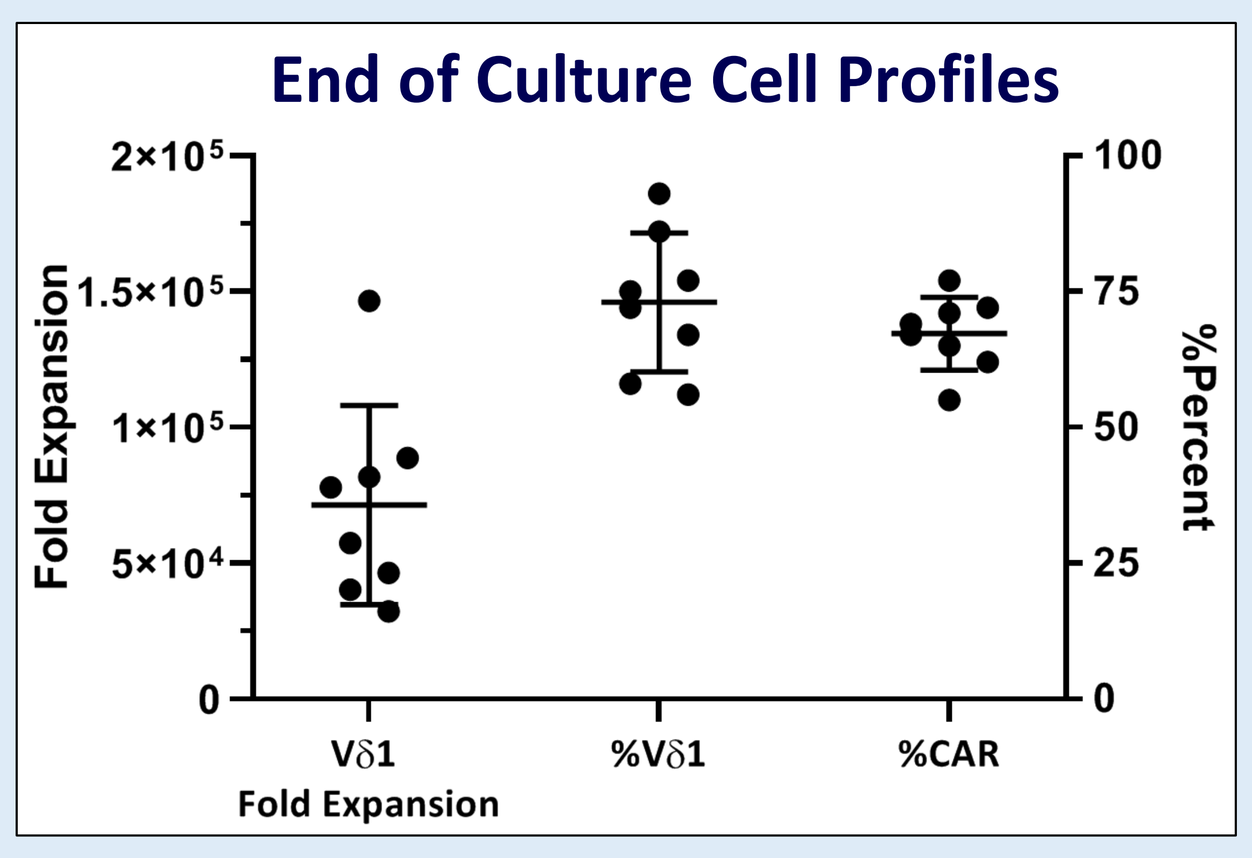

End of Culture Cell Profiles
Fold Expansion
%Percent
Vδ1
Fold Expansion
%Vδ1
%CAR

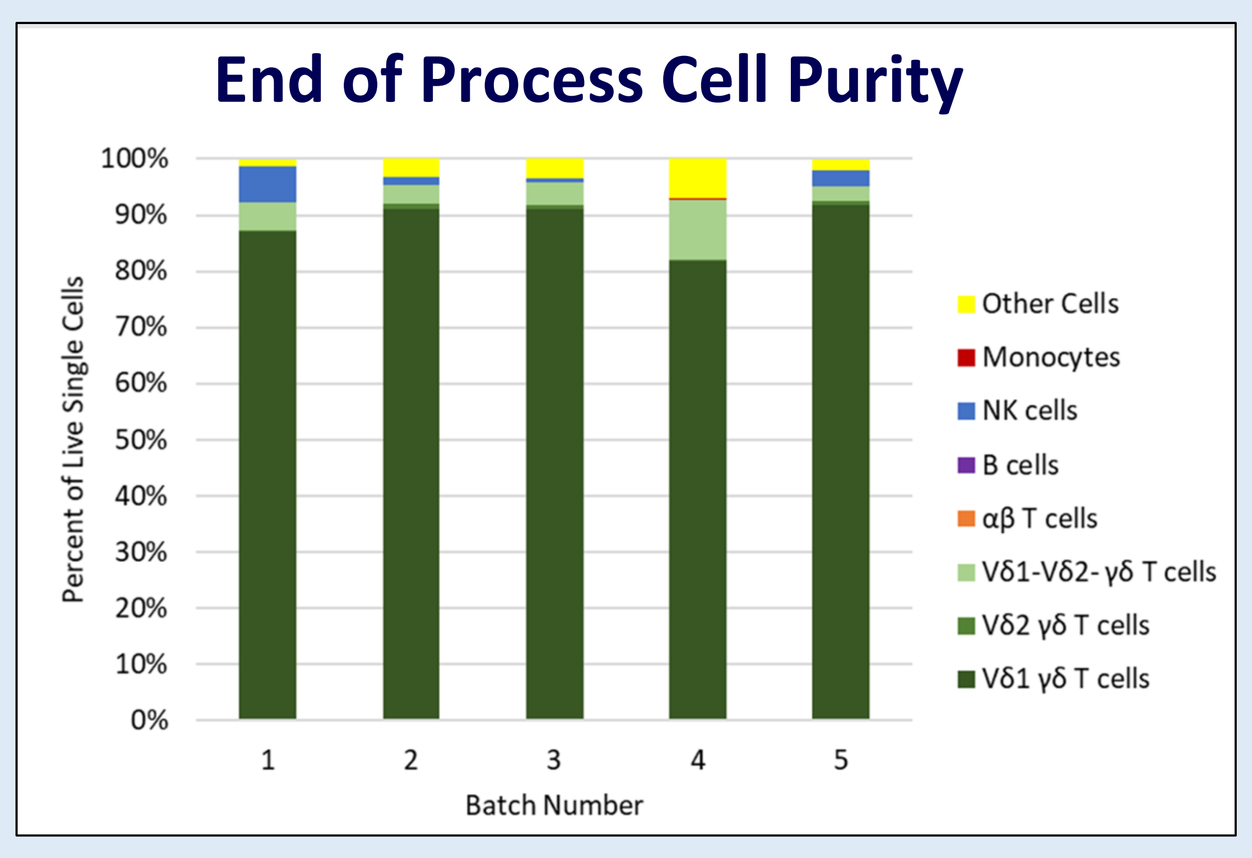

End of Process Cell Purity
Percent of Live Single Cells
Batch Number
Other Cells
Monocytes
NK cells
B cells
αβ T cells
Vδ1-Vδ2- γδ T cells
Vδ2 γδ T cells
Vδ1 γδ T cells

- **Healthy normal donors provide reliable and scalable source of PBMC starting material**
- **Donor screening identifies optimal donors that provide robust expansion, transduction and end of culture purity of Vδ1 CD20 CAR T cells**

Robust Cytotoxicity on B Cell Lymphoma Lines

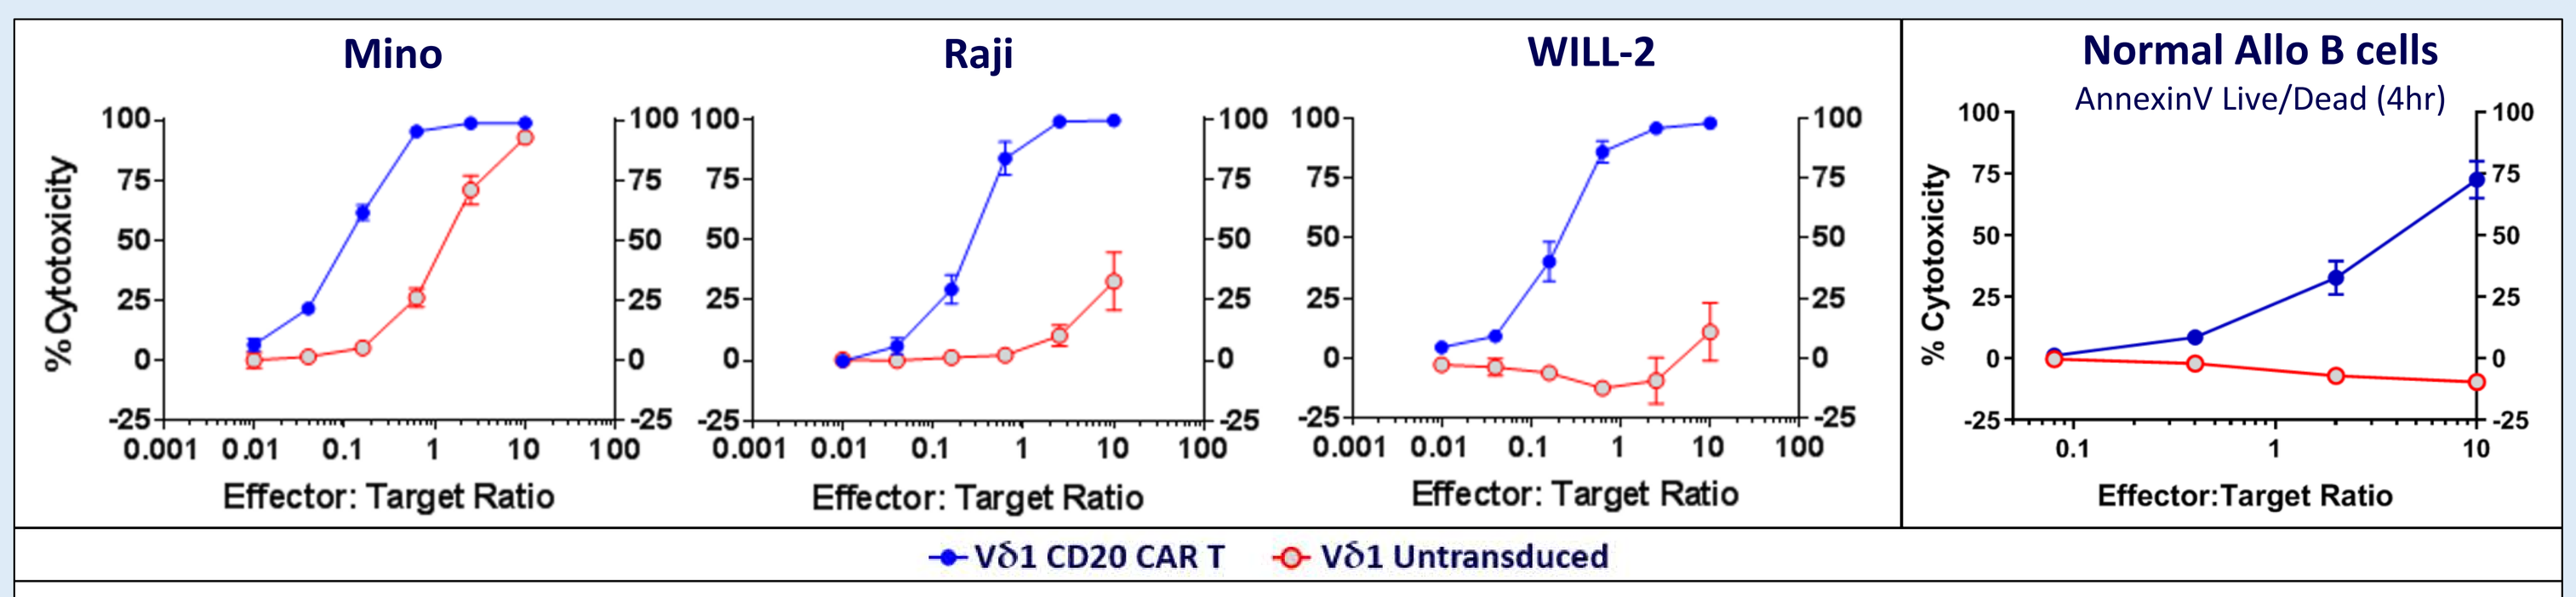

Mino
Raji
WILL-2
Normal Allo B cells
AnnexinV Live/Dead (4hr)
% Cytotoxicity
Effector: Target Ratio
Vδ1 CD20 CAR T
Vδ1 Untransduced

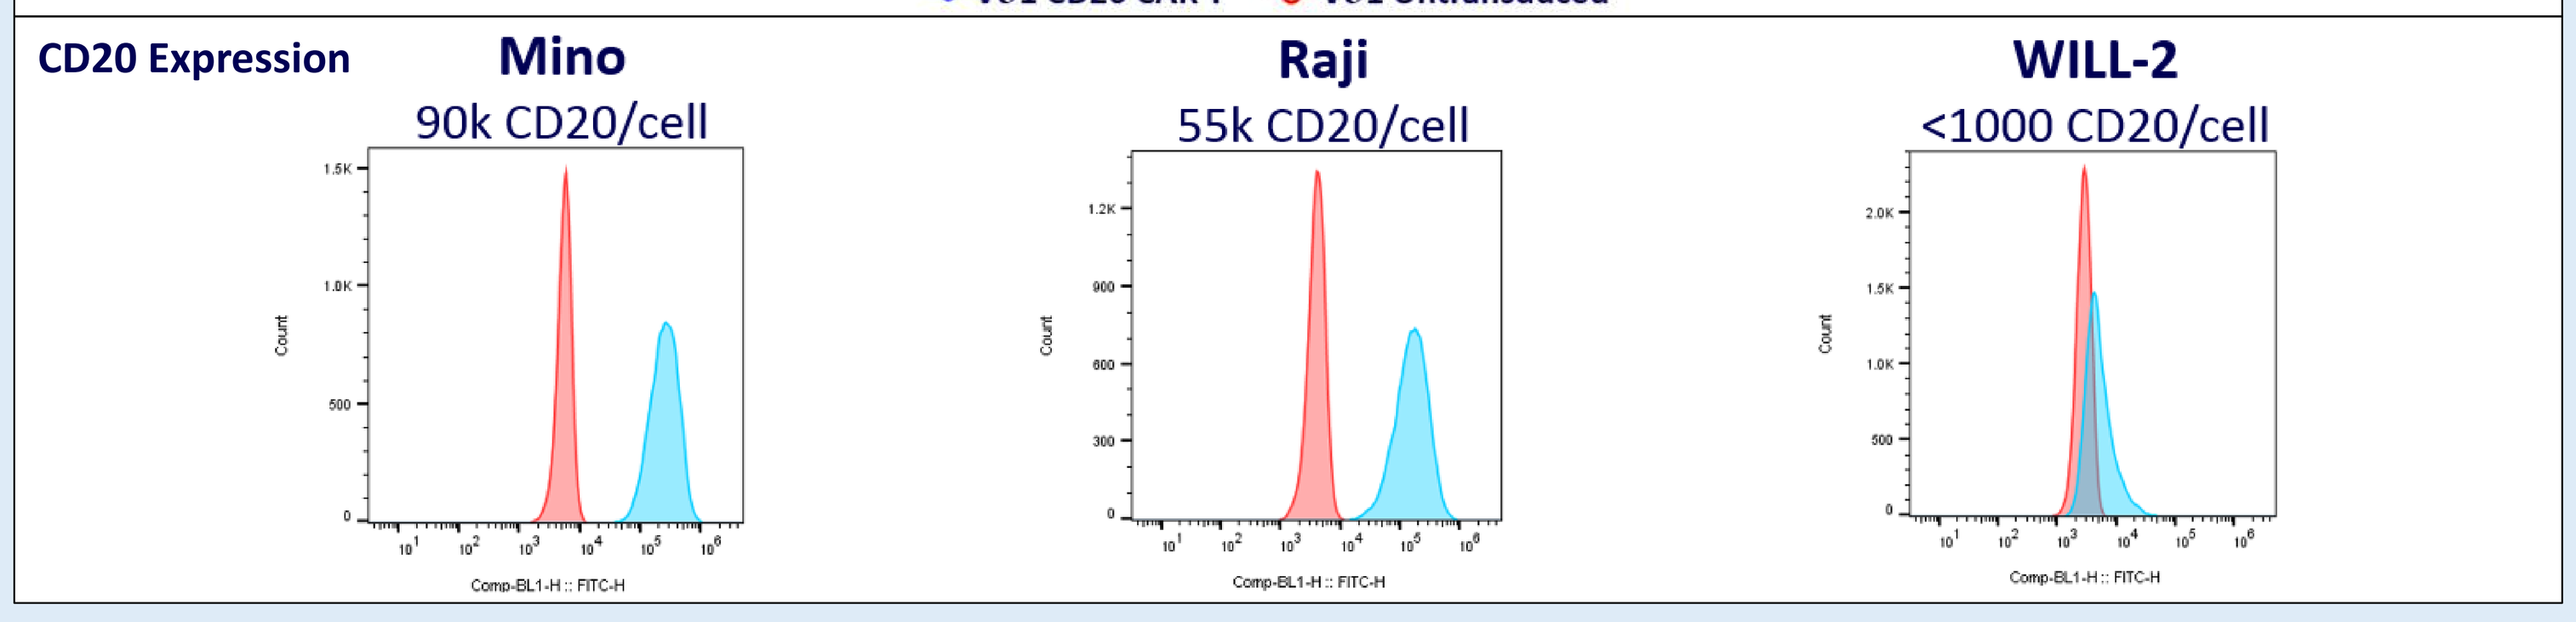

CD20 Expression
Mino
90k CD20/cell
Raji
55k CD20/cell
WILL-2
<1000 CD20/cell

Short-term cytotoxicity: Unengineered Vδ1 T cells show varying level of cytotoxicity on B cell lymphoma lines but not normal allogeneic B cells. Addition of CD20 CAR to Vδ1 T cells increases cytotoxic potency on CD20 expressing cells.

(Top panel): B cell lymphoma lines with luciferase reporters were co-incubated with expanded Vδ1 T cells for 18 hours, and luminescence was acquired. Values shown represent mean and standard deviation of %Cytotoxicity from three different donors. Normal B cells labeled with CFSE and co-incubated with expanded Vδ1 T cells for 4 hours. Viability was assessed by AnnexinV/DAPI. Values shown represent mean and standard deviation of %Cytotoxicity from four different donors.

(Bottom panel): CD20 expression level of B cell lymphoma lines as quantitated with Dako QifiKit.

- **Vδ1 CAR T cells show robust cytotoxic activity on CD20+ B cell lymphoma lines including WILL-2 (<10^3 CD20 copies/cell)**
- **Unengineered Vδ1 T cells show varying degree of innate cytotoxicity on wide range of cancer cell lines (no killing of Normal Allo B cells); introduction of CAR potentiates killing**

Persistent Cytotoxicity Potentiated by Exogenous IL-2

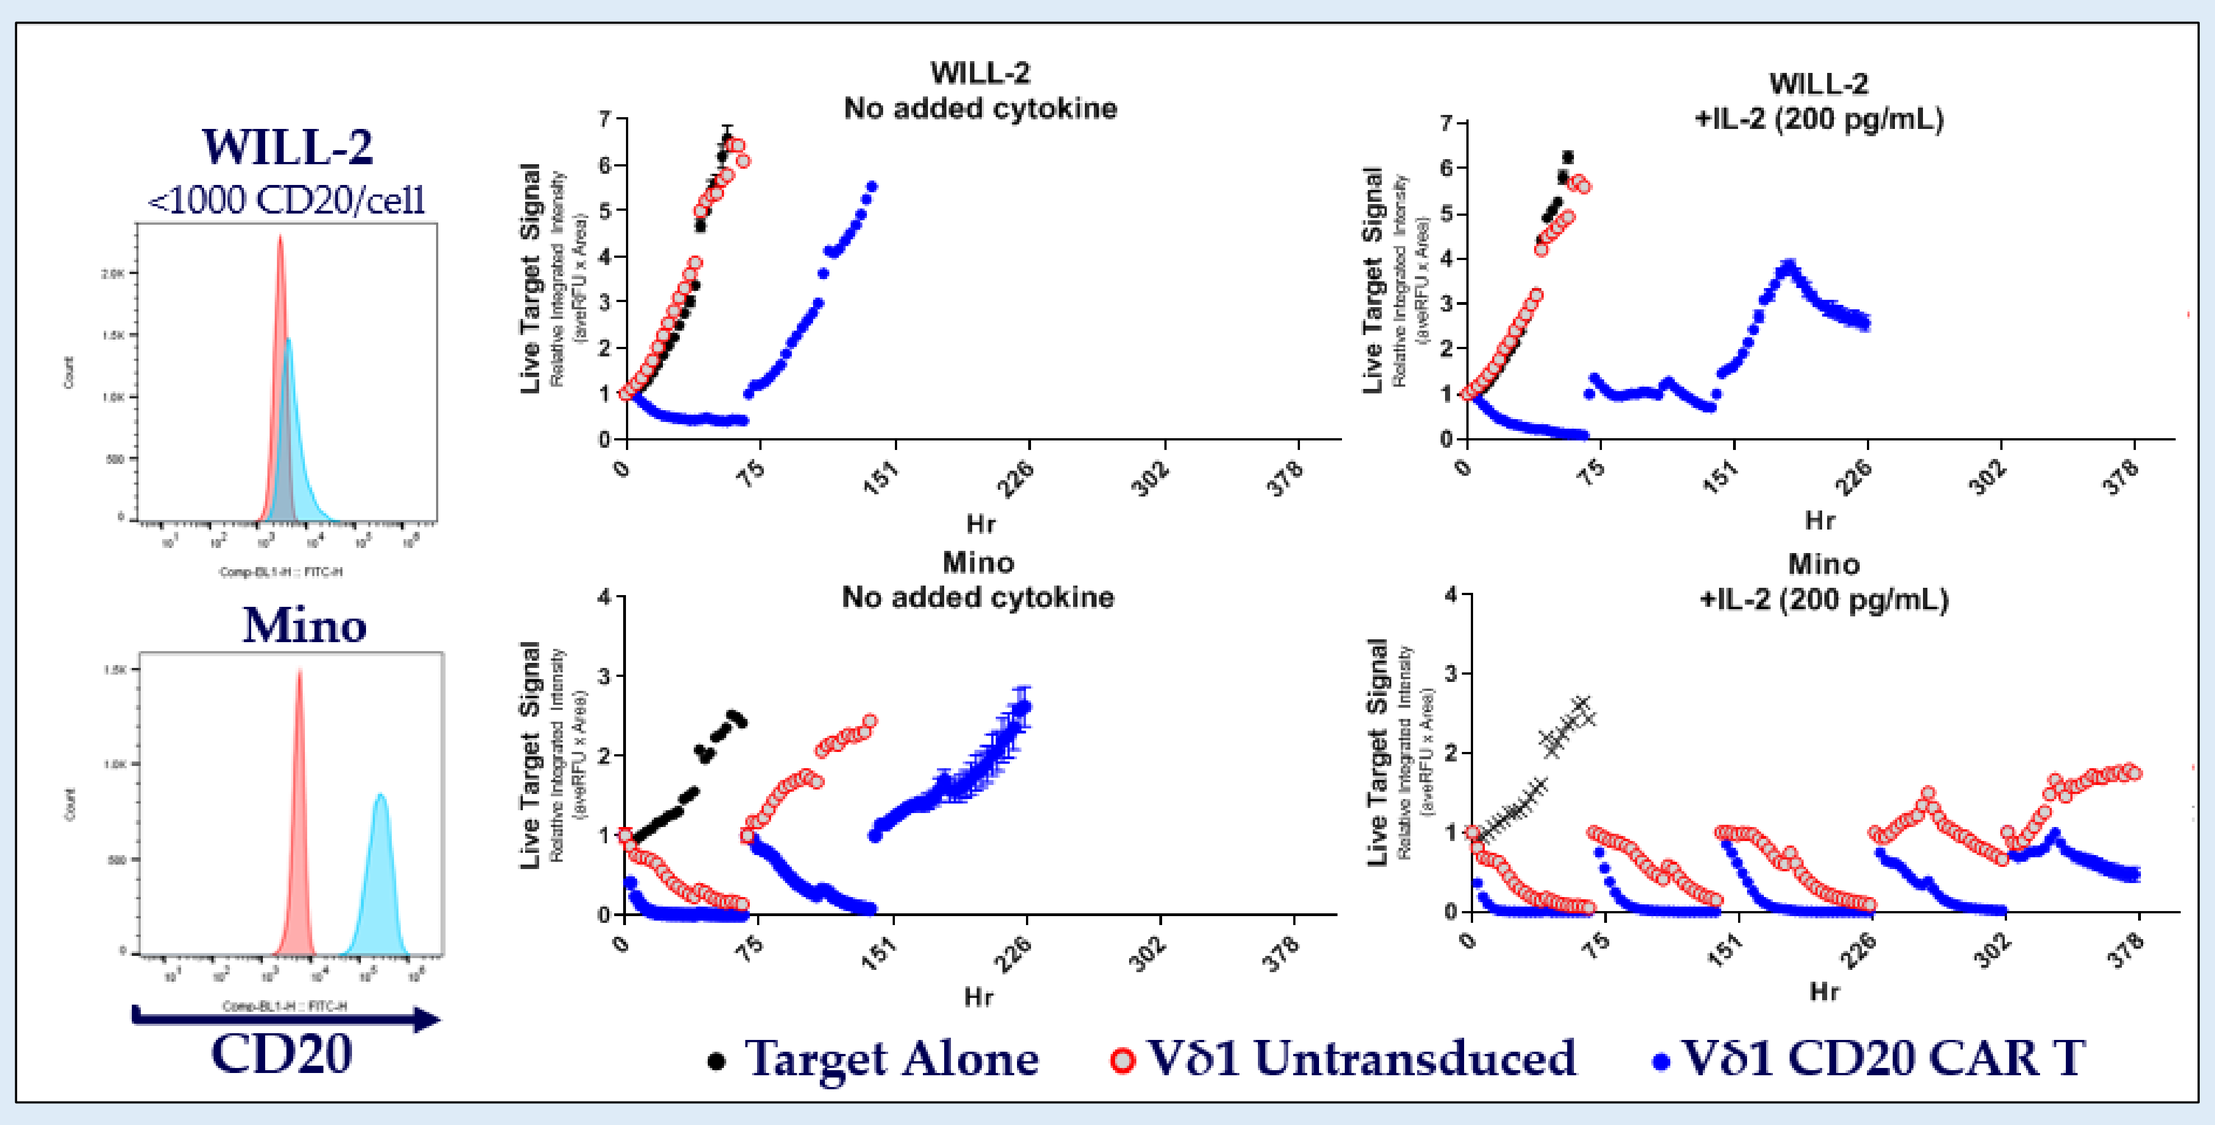

WILL-2
<1000 CD20/cell
Mino
CD20
WILL-2
No added cytokine
WILL-2
+IL-2 (200 pg/mL)
Mino
No added cytokine
Mino
+IL-2 (200 pg/mL)
Live Target Signal
Hr
Target Alone
Vδ1 Untransduced
Vδ1 CD20 CAR T

Donor ID	Description	% Viability Day1 Post-Thaw AnnexinV/DAPI	Mino 90k CD20/cell ~100 CD20/µm^2 +IL-2 200pg/mL	Mino -IL-2	Raji 55k CD20/cell ~100 CD20/µm^2 +IL-2 200pg/mL	Raji -IL-2	WILL-2 <1000 CD20/cell ~1.5 CD20/µm^2 +IL-2 200pg/mL	WILL-2 -IL-2	Jurkat CD20 Neg +IL-2 200pg/mL	Jurkat -IL-2
16	Vδ1 Untransduced	65	4.5	1.5	0.5	0.5	0	0	3.5	1
	Vδ1 CD20 CAR T	72	5	2.5	2.5	1.5	2.5	1	3.5	1
19	Vδ1 Untransduced	73	3.5	1.5	0.5	0.5	0	0	3.5	1
	Vδ1 CD20 CAR T	75	5	3.5	2.5	2.5	4.5	1	4.5	1.5
20	Vδ1 Untransduced	73	3.5	1.5	0.5	0.5	0	0	2.5	1
	Vδ1 CD20 CAR T	68	5	4.5	4.5	3.5	4.5	1.5	4.5	1

Effector Cell Information; Serial Killing Cytotox Results (Highest Round Cleared). E:T Ratio normalized to 3:1 based on viable CAR+ effectors

"Serial" Killing Assay: Longitudinal cytotoxicity performance of untransduced and CD20 CAR engineered Vδ1 T cells.

Initial ET ratio seeded at 3:1 of viable CAR+ Vδ1 T cells with 1.0e4 targets/well in a 384 well plate. Additional target cells are added at the end of each 72 hour "Round." Wells containing IL-2 (200pg/mL) are split 1:4 at the end of each 72 hour round, and wells without added IL-2 are split 1:2 at the end of each round beginning with the end of round two. Target signal is acquired and analyzed using Incucyte S3 Live-cell Analysis System.

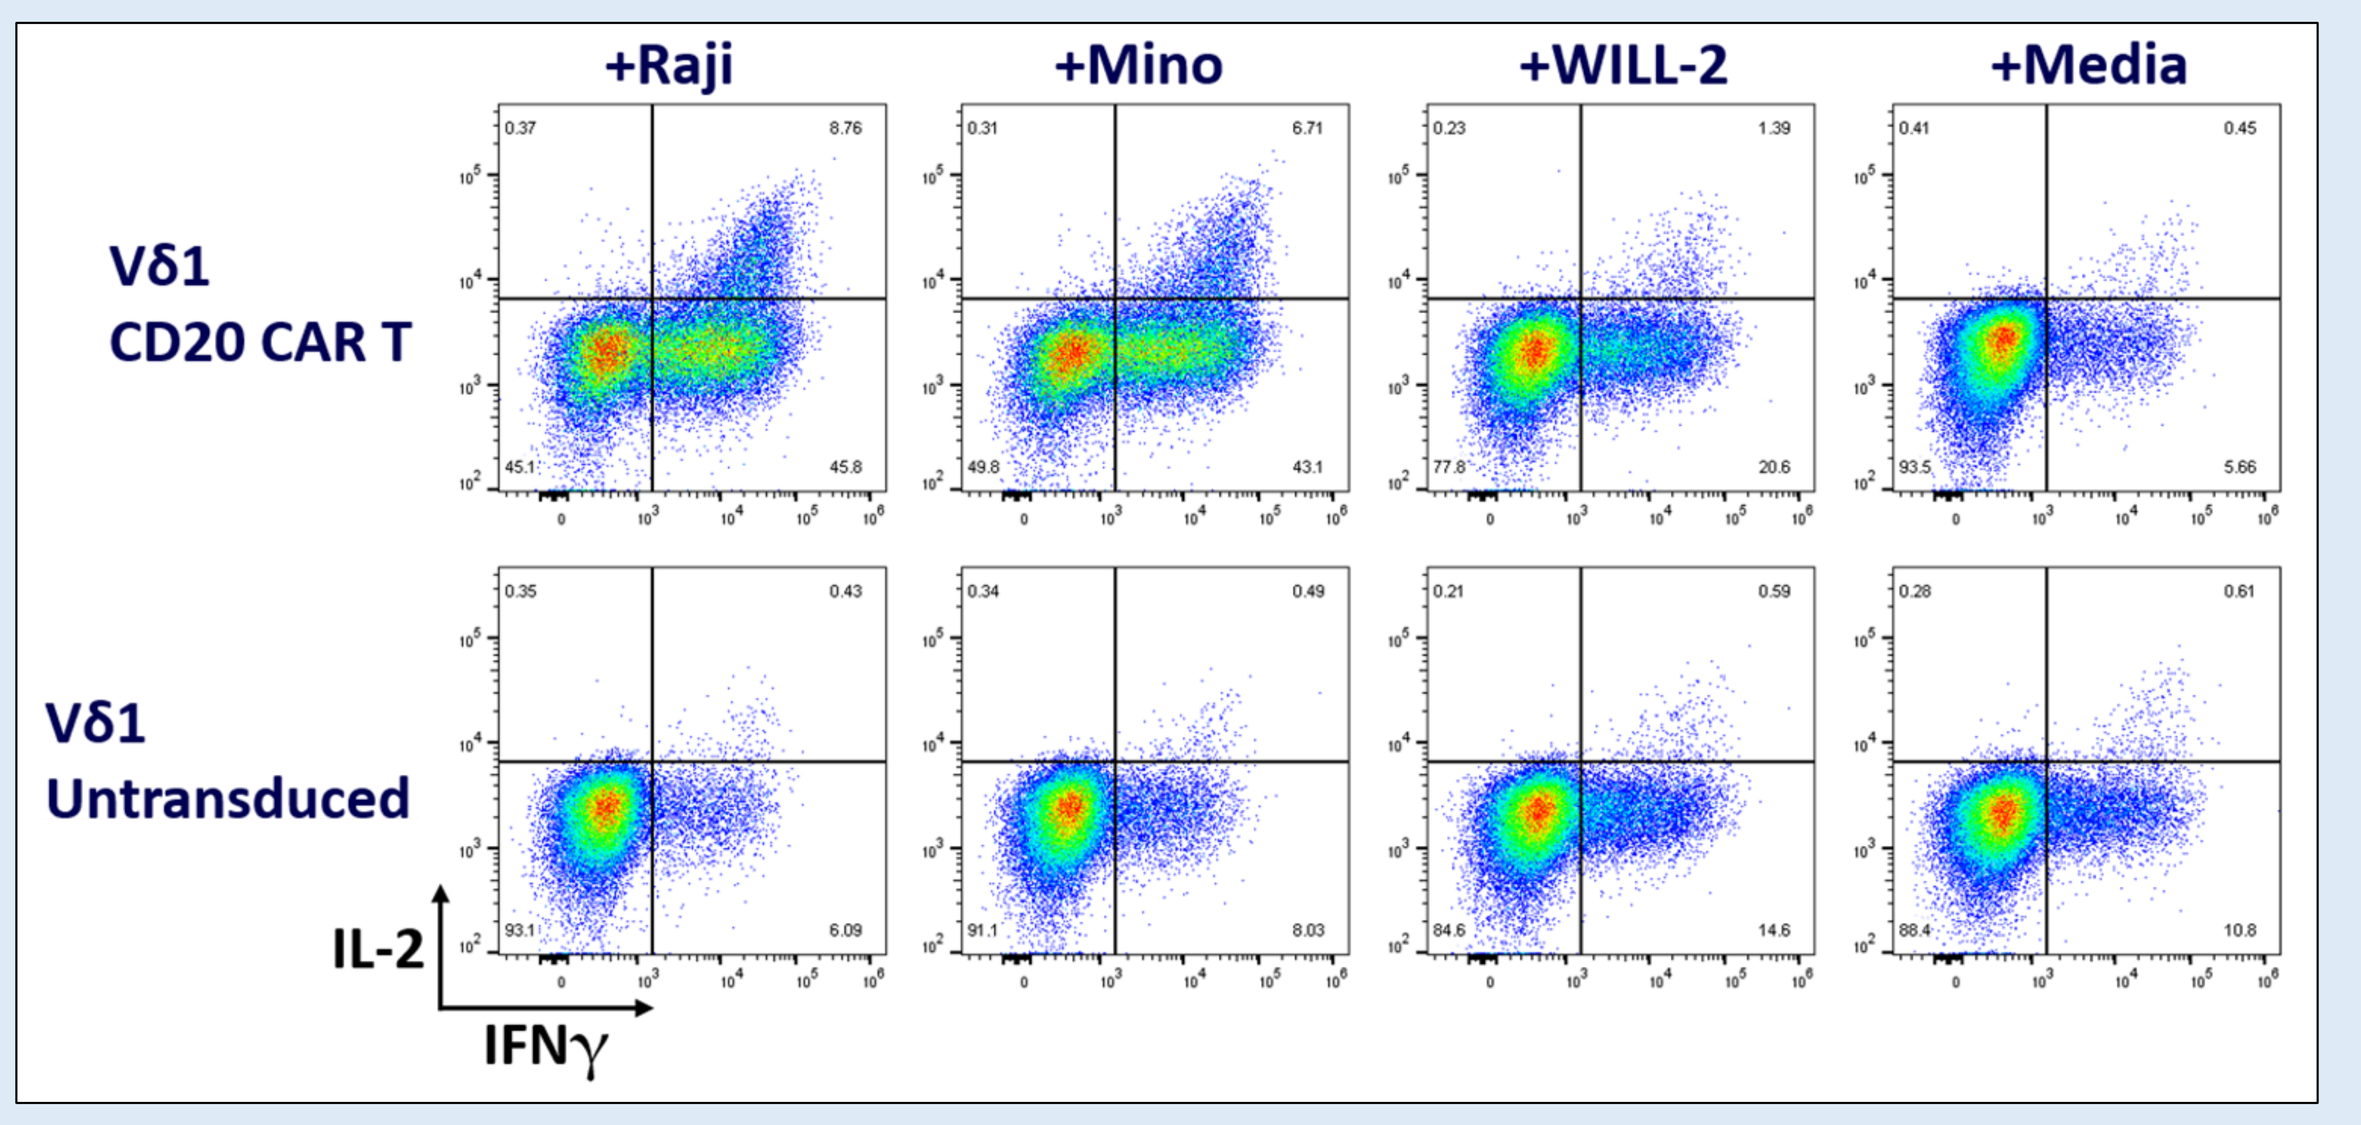

+Raji
+Mino
+WILL-2
+Media
Vδ1
CD20 CAR T
Vδ1
Untransduced
IL-2
IFNγ

Intracellular Cytokine Staining (ICS): Coculture of untransduced or CD20 CAR engineered Vδ1 T cells with CD20 expressing target cell lines elicits secretion of effector cytokines IFNγ and IL-2.

Vδ1 T cells and Target cells seeded at 1:1 ET Ratio in the presence of Brefeldin A. After 4 hours, cells were stained for cell surface Vδ1 TCR followed by intracellular staining of IL-2 and IFNγ. Only Vδ1 TCR+ population shown.

- **CAR engagement on Vδ1 T cells drives varying degree of cytokine secretion (notably IL-2) which depends partly on CAR target density**
- **Addition of IL-2 to cytotox coculture potentiates persistent innate cytotoxicity of untransduced Vδ1 T cells on wide range of cancer cell lines**
- **CAR mediated target recognition supports sustained cytotoxicity that is improved with addition of IL-2**

In Vivo Efficacy in Disseminated Lymphoma Tumor Models

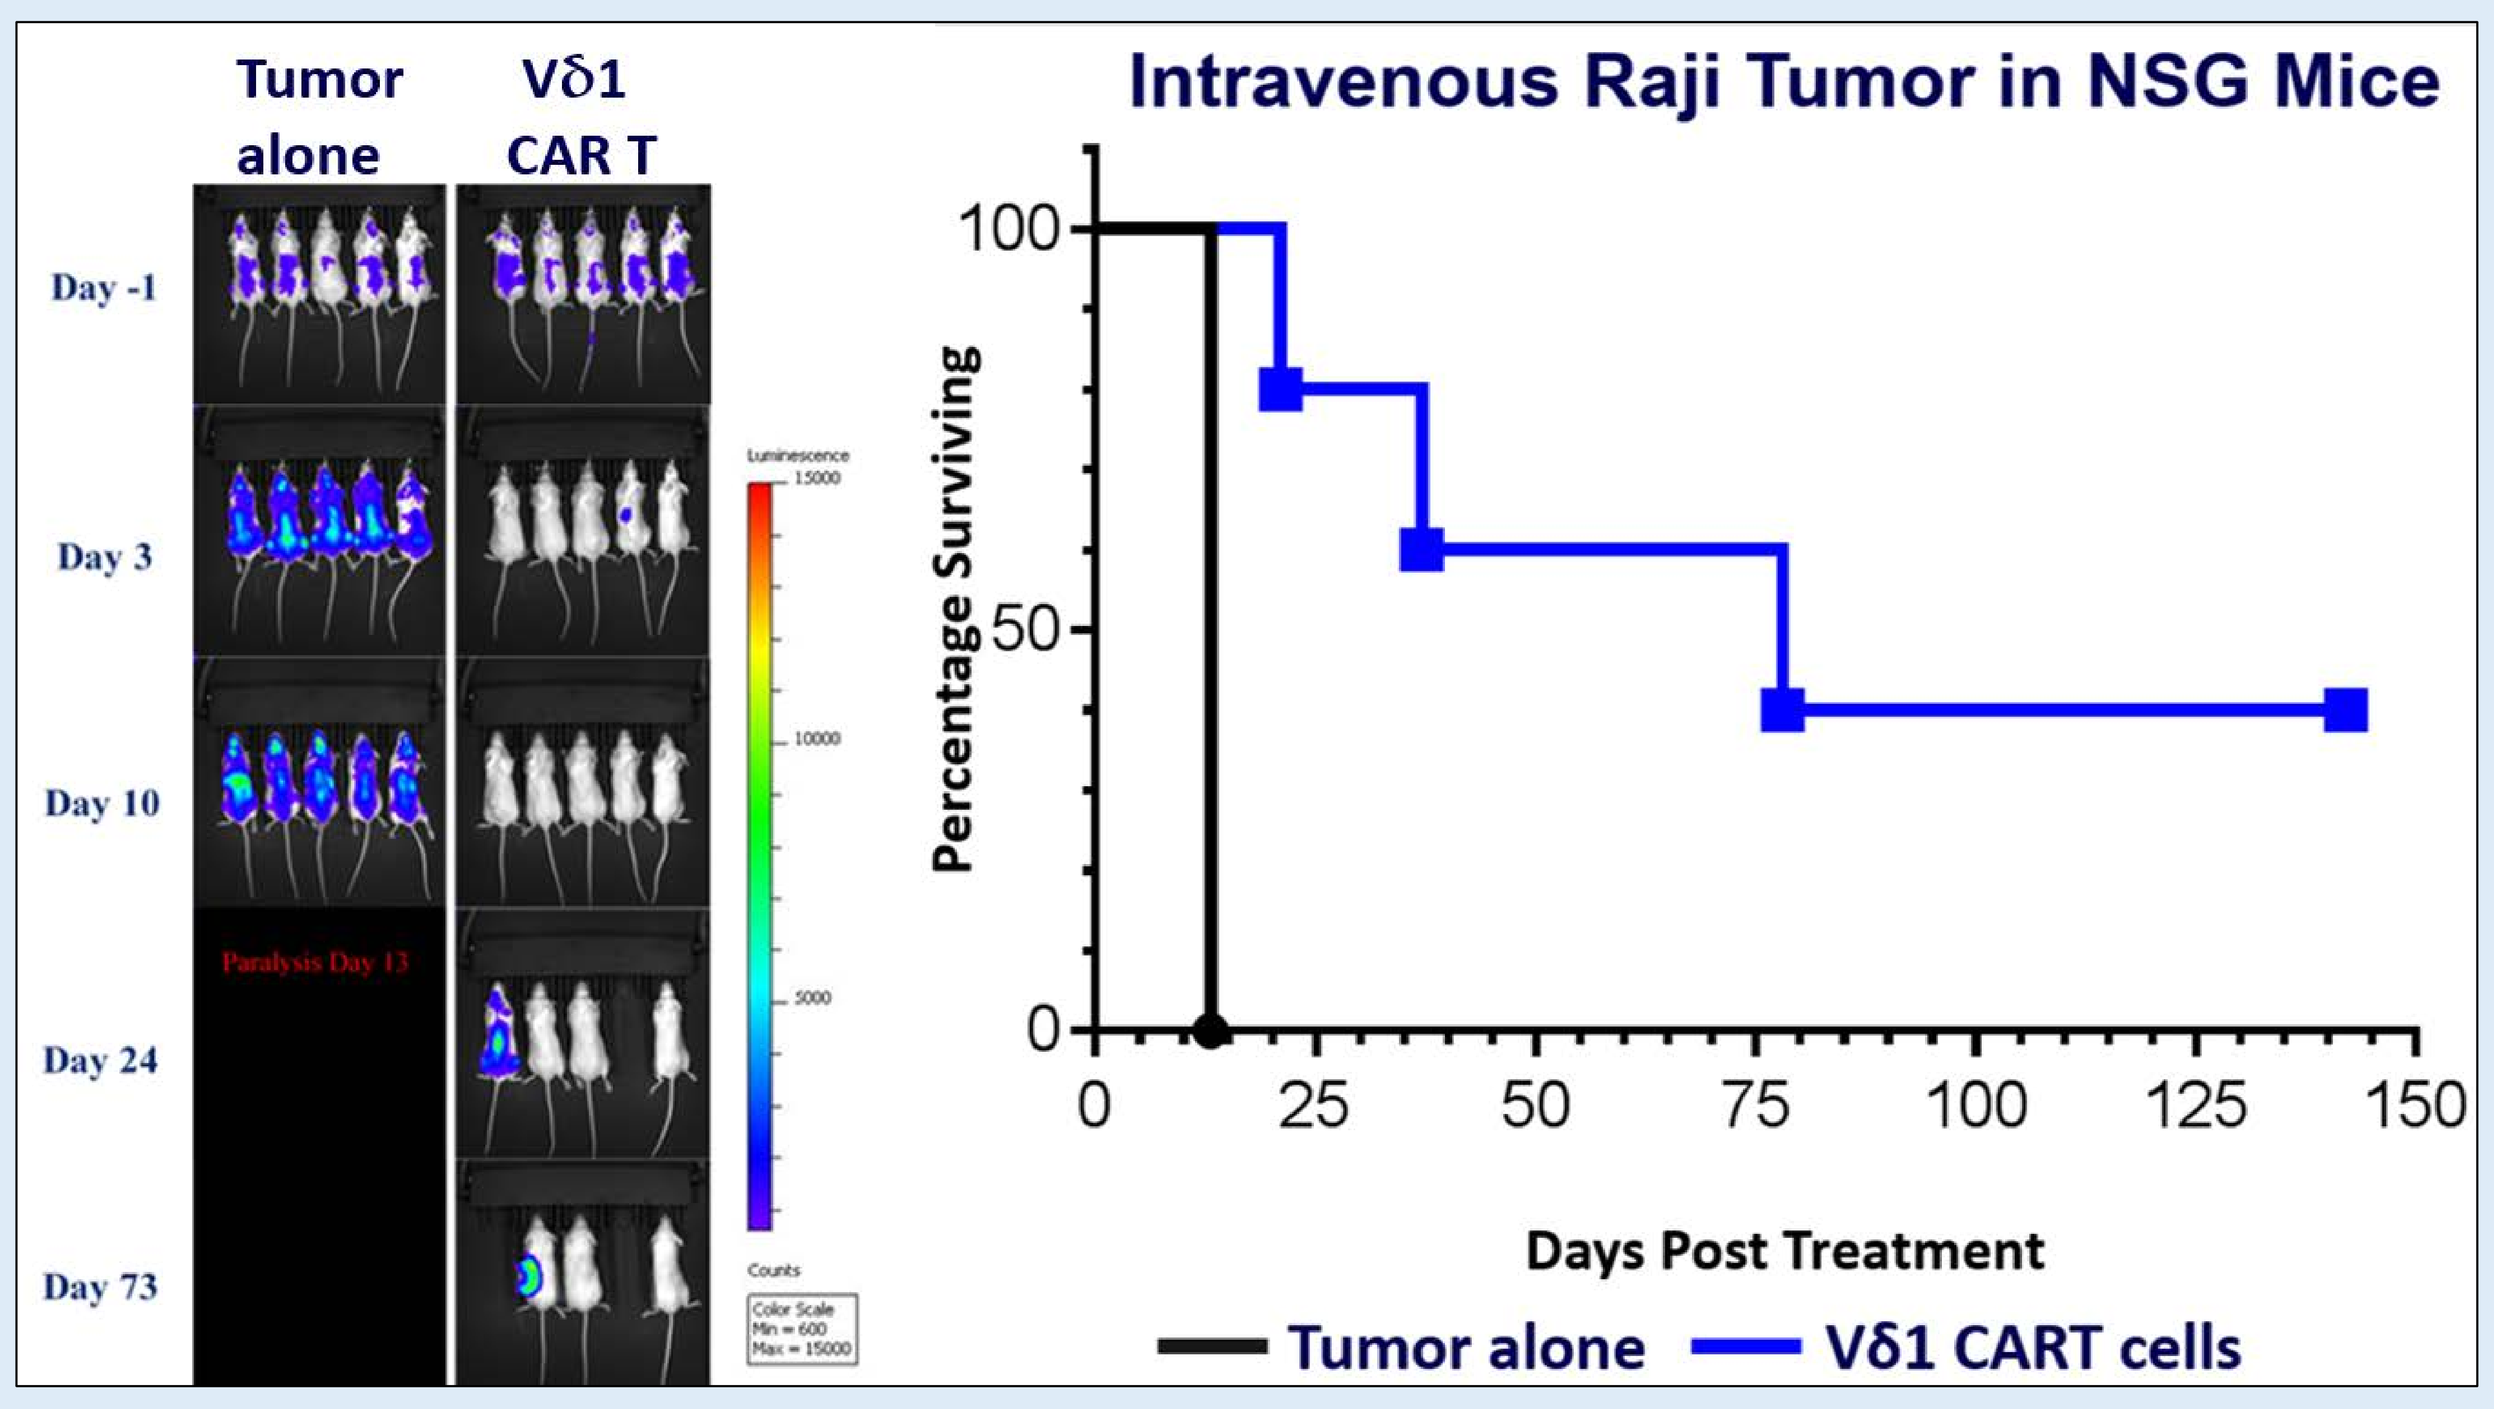

Tumor alone
Vδ1 CAR T
Day -1
Day 3
Day 10
Day 24
Day 73
Paralysis Day 13
Intravenous Raji Tumor in NSG Mice
Percentage Surviving
Days Post Treatment
Tumor alone
Vδ1 CART cells

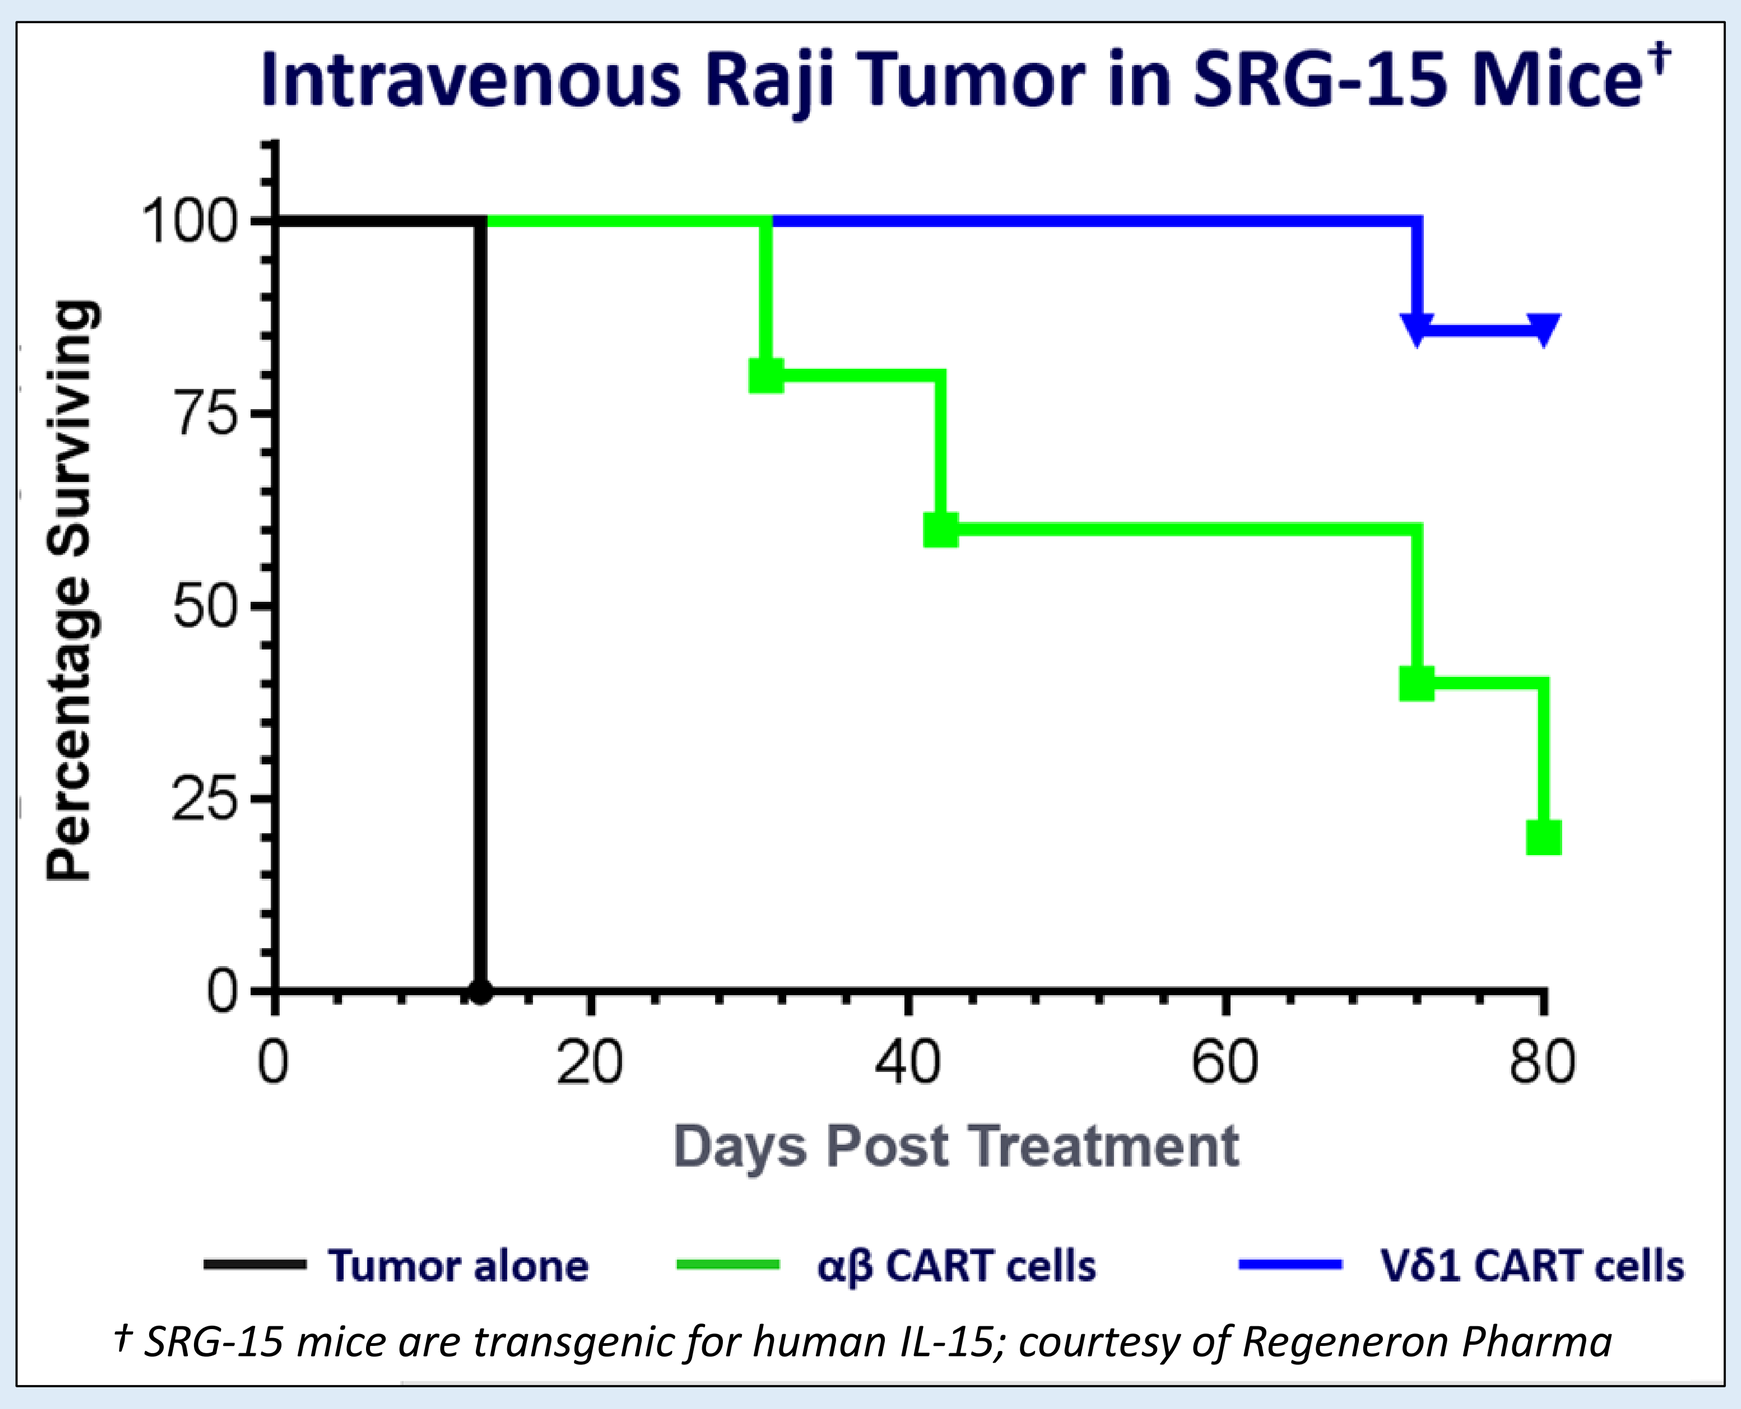

Intravenous Raji Tumor in SRG-15 Mice†
Percentage Surviving
Days Post Treatment
Tumor alone
αβ CART cells
Vδ1 CART cells
† SRG-15 mice are transgenic for human IL-15; courtesy of Regeneron Pharma

In Vivo Efficacy in Disseminated Tumor Model: Vδ1 CD20 CAR T cells mediate improved survival and tumor clearance in disseminated Raji tumor model that is supported by IL-2/IL-15.

(Left panel): NSG mice were implanted i.v. with 5e5 Raji-Luc cells. On day 4 post tumor implant, 9e6 Vδ1 CD20 CAR T cells were dosed i.v. Three doses of IL-2 supplementation were provided at a dose of 13,000 IU s.c. at 1 hour pre-treatment and day 1 and day 2 post treatment. n=5 mice per group. **(Right panel):** SRG-15 mice transgenic for human IL-15 (Herndler-Brandstetter et al., 2017, PNAS) were implanted i.v. with 5e5 Raji-Luc cells. On day 4 post tumor implant, 2e7 Vδ1 CD20 CAR T cells or 7e6 αβ CAR T cells were dosed i.v. n=7 mice per group. For both studies, survival was observed with humane endpoints of ≥ 20% loss of initial body weight and/or onset of partial or full paralysis.

- **Treatment with Vδ1 CD20 CAR T in aggressive disseminated Raji tumor model leads to tumor clearance, growth inhibition and improved survival**
- **No GvHD observed in animals treated with Vδ1 CD20 CAR T cells whereas αβ CD20 CAR T cells induce GvHD (SRG-15 mice)**
- **No gene editing of Vδ1 T cells required to overcome GvHD**

In Vivo Efficacy of Vδ1 CD20 CAR T Cells Improved with Addition of Secreted IL-15

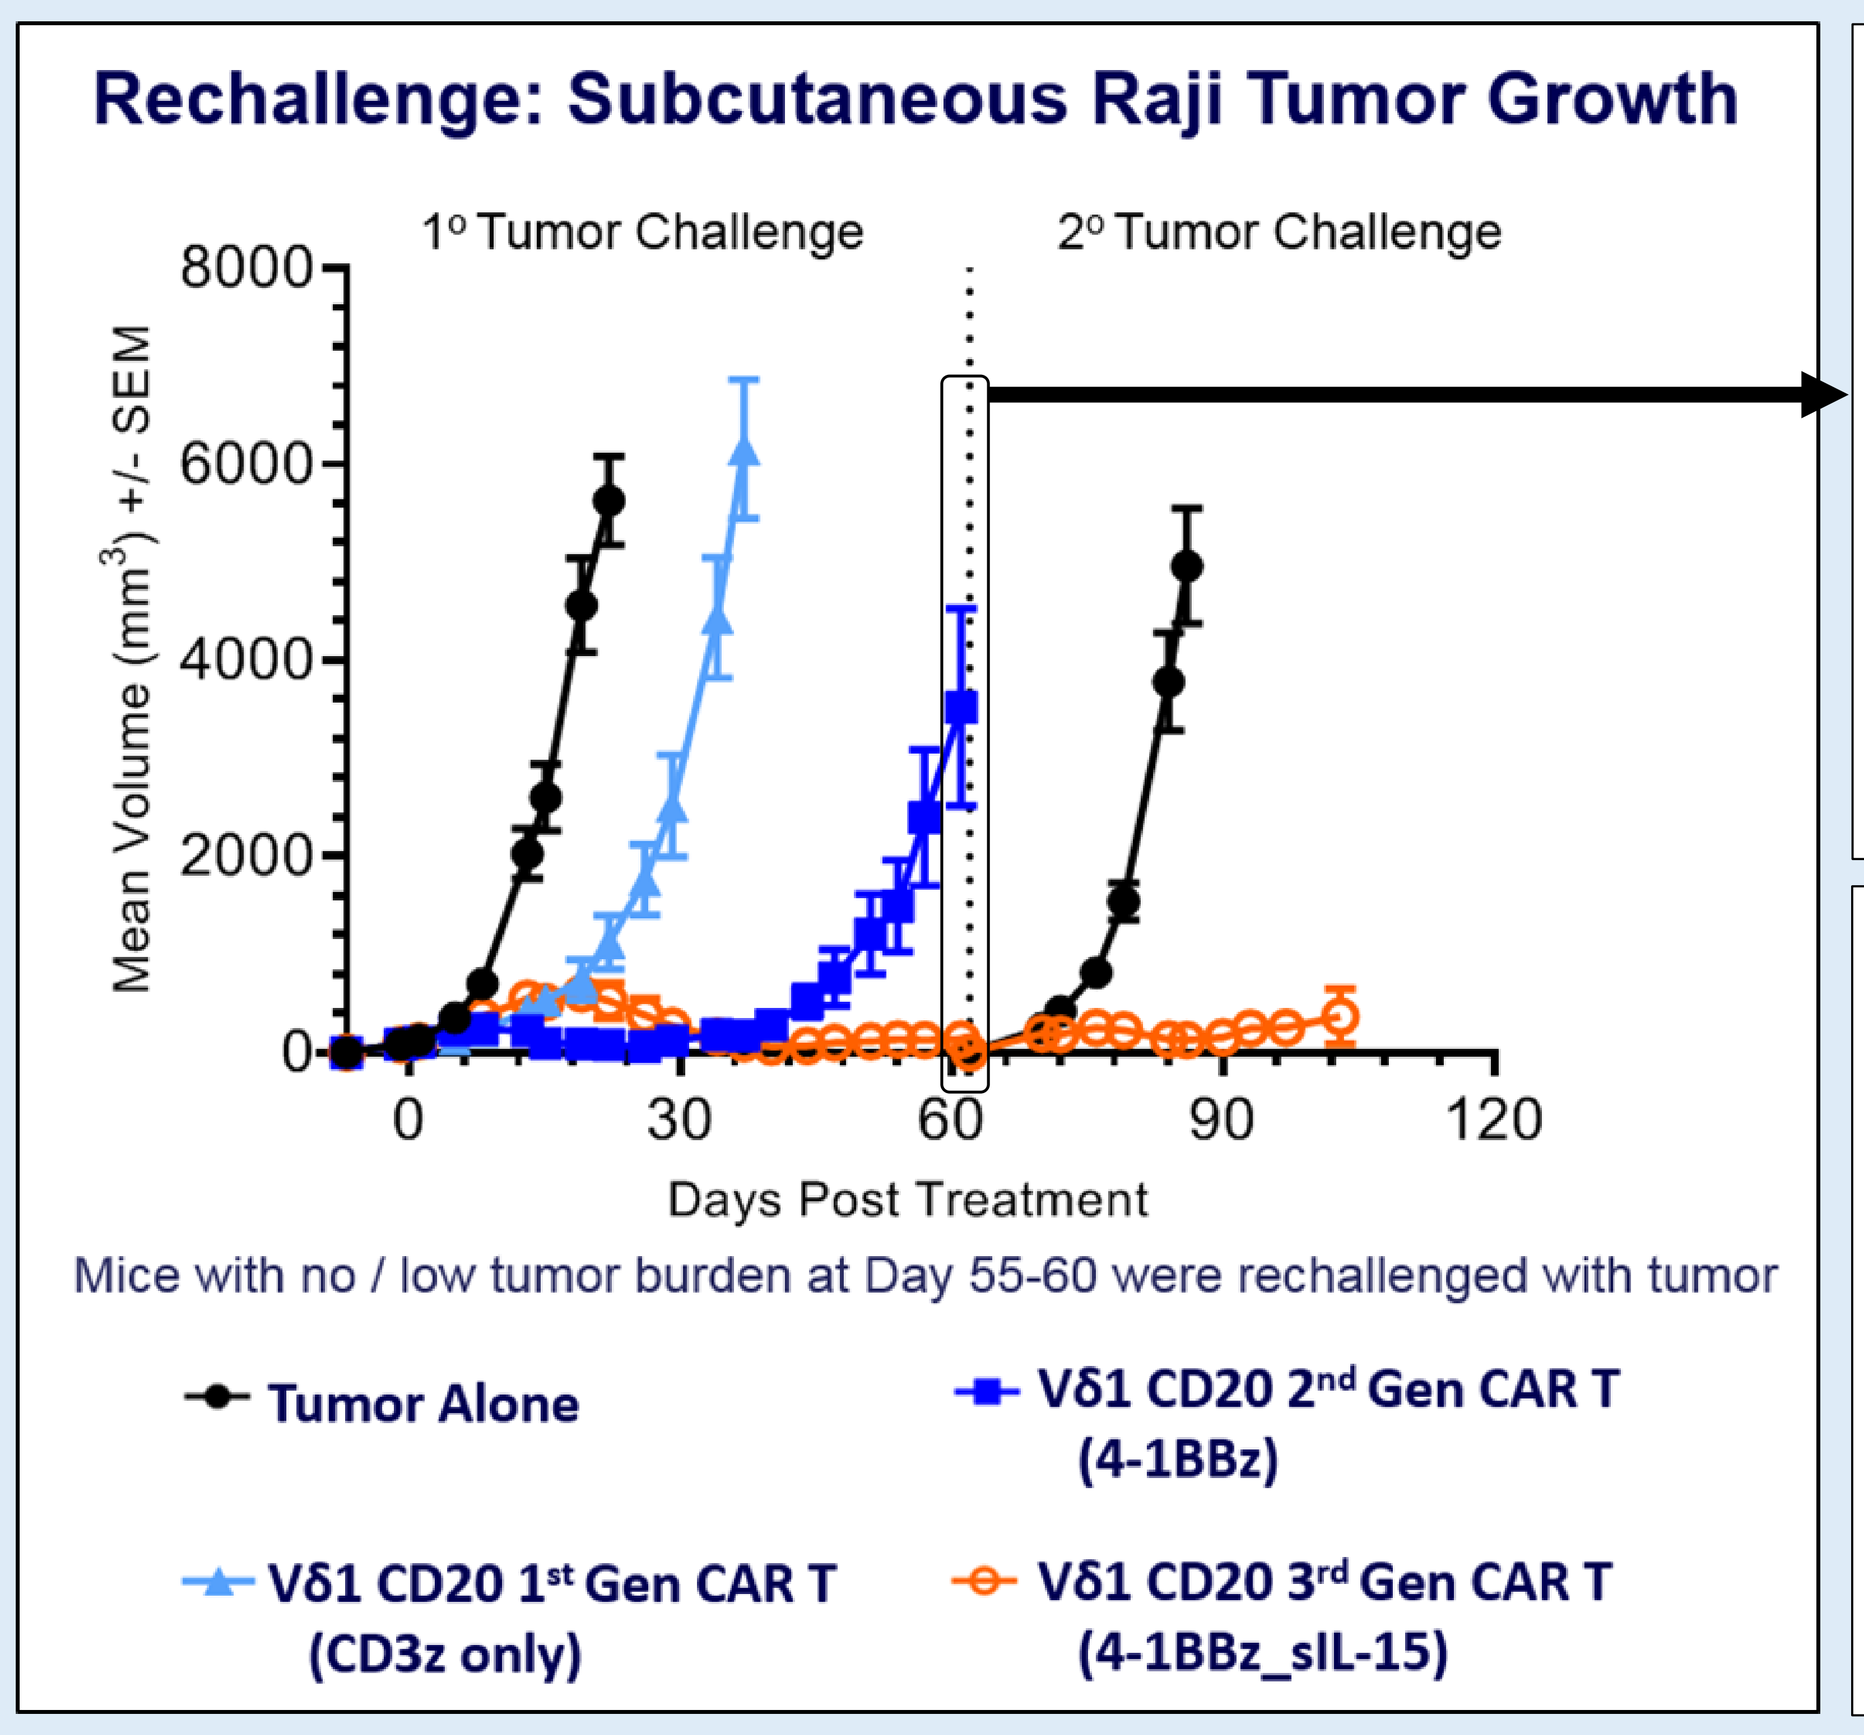

Rechallenge: Subcutaneous Raji Tumor Growth
1° Tumor Challenge
2° Tumor Challenge
Mean Volume (mm³) +/- SEM
Days Post Treatment
Mice with no / low tumor burden at Day 55-60 were rechallenged with tumor
Tumor Alone
Vδ1 CD20 2nd Gen CAR T (4-1BBz)
Vδ1 CD20 1st Gen CAR T (CD3z only)
Vδ1 CD20 3rd Gen CAR T (4-1BBz_sIL-15)

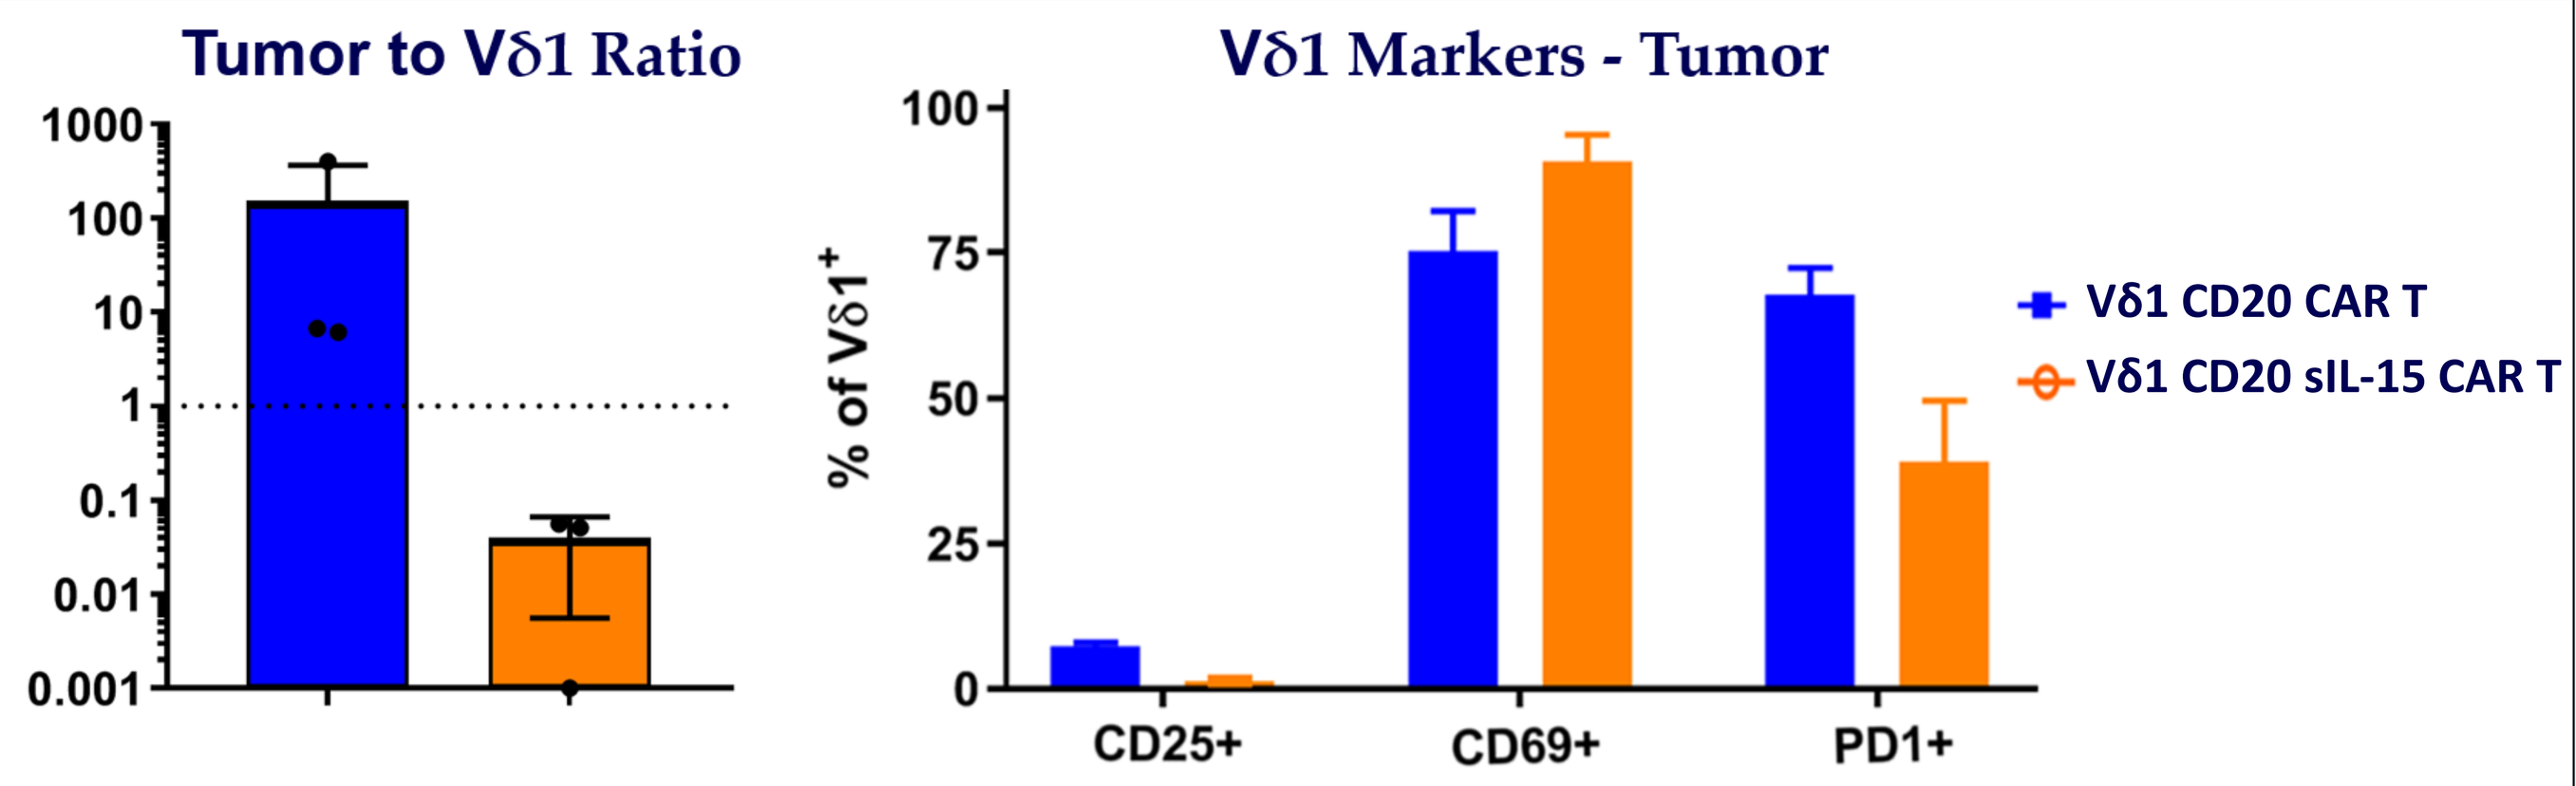

Tumor to Vδ1 Ratio
Vδ1 Markers - Tumor
% of Vδ1+
CD25+
CD69+
PD1+
Vδ1 CD20 CAR T
Vδ1 CD20 sIL-15 CAR T

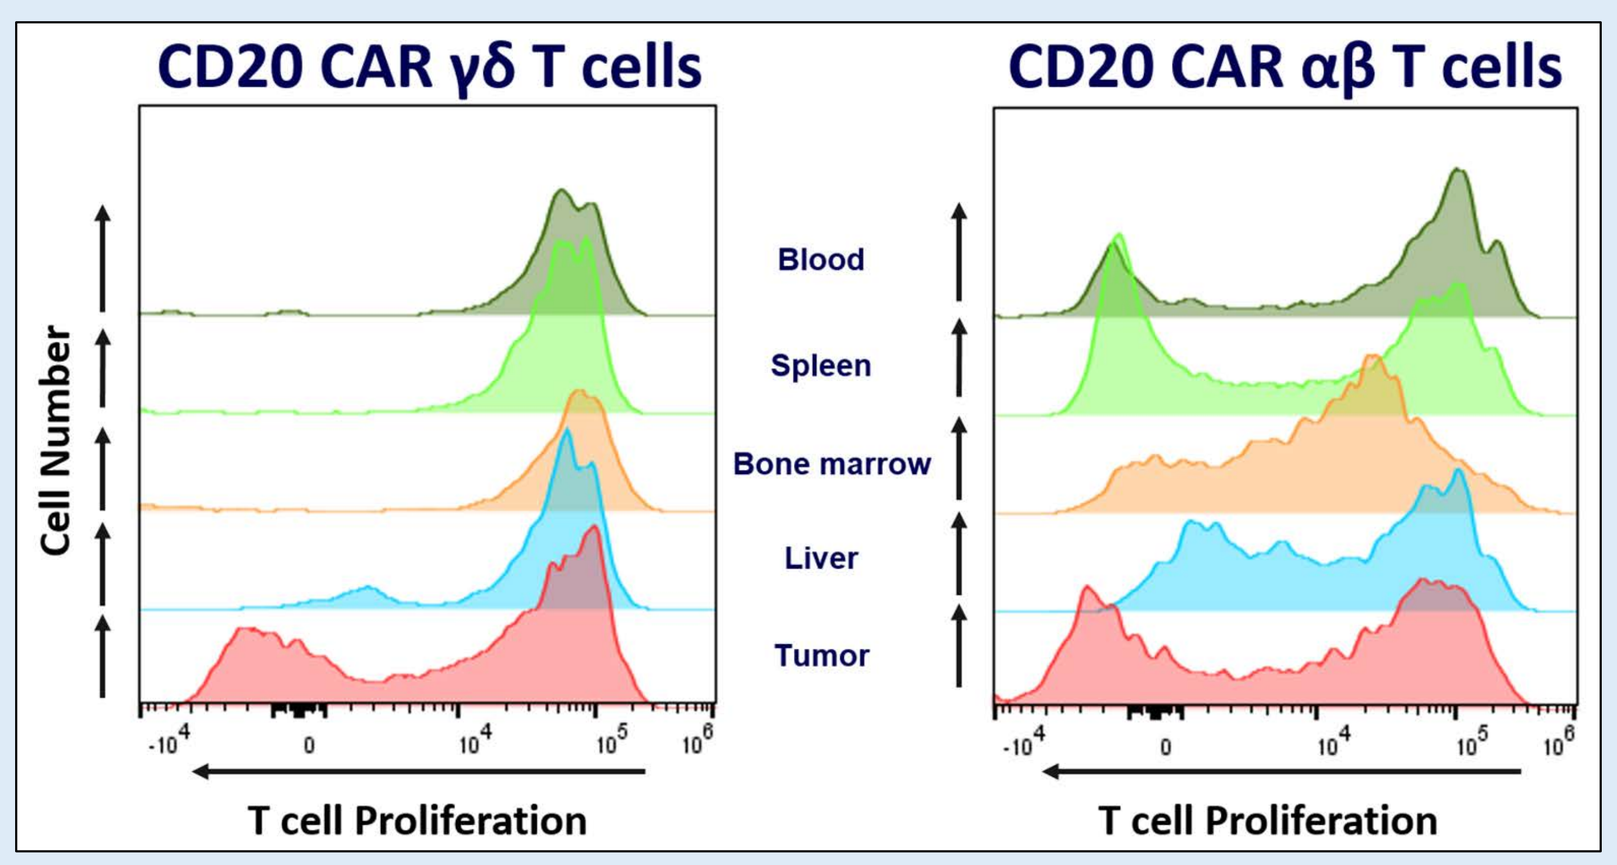

CD20 CAR γδ T cells
CD20 CAR αβ T cells
Cell Number
Blood
Spleen
Bone marrow
Liver
Tumor
T cell Proliferation

Ex Vivo Analysis of CAR T cell proliferation: Vδ1 CD20 CAR T cells show proliferation in tumor whereas αβ CD20 CAR T cell show proliferation in all organs tested.

Satellite groups of mice from the subcutaneous efficacy study shown were sacrificed at day 6 post T cell implant and organs were dissociated. Cell-Trace-Violet labeling of Vδ1 and αβ CD20 CAR T cells prior to implant enabled assessment of proliferation by dye dilution.

In Vivo Efficacy in Subcutaneous Tumor Model: Vδ1 CD20 CAR T cells control subcutaneous Raji tumors with improved efficacy observed with 4-1BB signaling domain and secreted IL-15

(Left panel): NSG mice were implanted subcutaneously with 1e6 Raji cells. On day 7 post tumor implant, 5e6 of indicated Vδ1 CD20 CAR T cells were dosed i.v., n=7 mice per group. IL-2 supplementation was provided in doses of 13,000 IU i.p. at 1 hour pre-treatment and then three times per week for the duration of the study. Tumor Rechallenge with subcutaneous implant of 1e6 Raji cells at day 60 in mice with little/no residual tumor (n=4 mice) was performed to assess persistence of IL-15 secreting Vδ1 CD20 CAR T cells. **(Top-right panel):** At day 60 post treatment, tumors were excised and dissociated for FACS assessment of Raji tumor to Vδ1 T cell ratio and Vδ1 T cell markers CD25, CD69 and PD-1 (n=3 mice).

- **Improved control of subcutaneous Raji tumors by Vδ1 CD20 CAR T cells attributable to 4-1BB signaling domain**
- **Next generation construct with addition of secreted IL-15 shows further improvement in efficacy**
 - **Tumor-free animals continue to show tumor growth control upon re-challenge with more tumor cells at Day 60**
- **Tumor specific proliferation of Vδ1 CD20 CAR T cells observed versus multi organ proliferation of αβ CD20 CAR T cells in NSG mice**
- **Majority of long-term, tumor-resident Vδ1 CAR T cells are PD-1+**
 - **CAR expression may be downregulated; cells are functional if extracted from tumor, rested overnight, and assessed ex vivo**
 - **Combination treatment with PD1 inhibitor is being planned**

Summary

- **Healthy donor-derived CD20 CAR-engineered Vδ1 cells can be consistently manufactured to create an 'off-the-shelf' immune cell product**
- **Vδ1 CD20 CAR T cells display robust anti-tumor activity against a range of CD20 expressing tumor lines in vitro and demonstrate potent effector cytokine production and upregulation of activation markers (e.g. CD69)**
- **Vδ1 CD20 CAR T cells expand in response to target cell engagement and expansion is augmented by exogenous homeostatic cytokines, such as IL-2 or IL-15, both in vitro and in vivo**
- **Vδ1 CD20 CAR T cells show robust in vivo activity in disseminated and s.c. models of B cell lymphomas**
- **Vδ1 CAR T cells do not induce GvHD in murine models**
- **Large-scale cGMP-compliant manufacture of healthy donor-derived Vδ1 CD20 CAR T cells is expected to provide a safe and effective therapeutic option for patients with CD20 expressing malignancies; clinical studies are anticipated to start in early 2020**